Abstract 8589

Depth of response and progression-free survival in patients with advanced *ALK*-positive non-small cell lung cancer treated with lorlatinib

Rosario García Campelo,[1] Christian Schulz,[2] Young Joo Min,[3] Adnan Aydiner,[4] Maen Hussein,[5] Alessandra di Pietro,[6] Anna Polli,[6] Jean-François Martini,[7] Hye Ryun Kim[8]

[1]University Hospital A Coruña, A Coruña, Spain; [2]University Hospital Regensburg, Regensburg, Germany; [3]University of Ulsan College of Medicine, Ulsan, Republic of Korea; [4]Istanbul University, Institute of Oncology, Istanbul, Turkey; [5]Florida Cancer Specialists & Research Institute, Lady Lake, FL, USA; [6]Pfizer, Milan, Italy; [7]Pfizer, La Jolla, CA, USA; [8]Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Republic of Korea

Conclusions

- With lorlatinib treatment, 80% of patients experienced >50% shrinkage in target lesion, and 34% had >75% shrinkage in target lesion
- Greater depth of response (DepOR) was associated with longer progression-free survival (PFS) in patients with advanced anaplastic lymphoma kinase (*ALK*)-positive non-small cell lung cancer (NSCLC) treated with lorlatinib
 - The probability of remaining progression free at 5 years was 75% in patients with DepOR of >75%-100%, compared with 37% in patients with DepOR of 0%-50%
- No differences were observed in *ALK* fusion variants in the DepOR groups
- No association was observed between DepOR and circulating tumor DNA (ctDNA) clearance status

Electronic Poster

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster. If you don't have a smartphone, access the poster via the internet at: https://scientificpubs.congressposter.com/p/ng42i2c6ouvja0q5

Abstract Plain Language Summary

Please scan this QR code with your smartphone to view a plain language summary of the abstract. If you do not have a smartphone, access the summary via the internet at: https://scientificpubs.congressposter.com/pls/ng42i2c6ouvja0q5

Correspondence: Rosario García Campelo, ma.rosario.garcia.campelo@sergas.es

References: 1. Lorbrena (lorlatinib). Prescribing information. Pfizer; 2024. Accessed March 19, 2025. http://labeling.pfizer.com/ShowLabeling.aspx?id=11140. 2. Shaw AT, et al. *N Engl J Med*. 2020;383:2018-2029. 3. Solomon BJ, et al. *J Clin Oncol*. 2024;42:3400-3409. 4. McCoach CE, et al. *Ann Oncol*. 2017;28:2707-2714.

Acknowledgments: The authors thank the participating patients and their families, investigators, subinvestigators, research nurses, study coordinators, and operations staff. During the preparation of this work the authors used a genAI tool (03/17/25; Pfizer; GPT-4o) to develop the first draft. After using this tool/service, the authors reviewed and edited the content as needed and take full responsibility for the content of the publication. Editorial assistance was provided by Kakoli Parai, PhD, of Nucleus Global, and funded by Pfizer.

Disclosures: Dr. García Campelo reports honorarium from Amgen, AstraZeneca, Bayer, BMS, Boehringer Ingelheim, F. Hoffmann-La Roche, Janssen, Lilly, MSD, Pfizer, Sanofi, and Takeda; consulting or advisory role at Amgen, AstraZeneca, Bayer, BMS, Boehringer Ingelheim, F. Hoffmann-La Roche, Janssen, Lilly, MSD, Pfizer, Sanofi, and Takeda; participating in speakers' bureau for Amgen, AstraZeneca, Bayer, BMS, Boehringer Ingelheim, F. Hoffmann-La Roche, Janssen, Lilly, MSD, Pfizer, Sanofi, and Takeda; and research funding from Amgen, AstraZeneca, Bayer, BMS, Boehringer Ingelheim, F. Hoffmann-La Roche, Janssen, Lilly, MSD, Pfizer, Sanofi, and Takeda.

Presented at the American Society of Clinical Oncology Annual Meeting 2025 | May 30-June 3, 2025 | Chicago, IL, USA

Background

- Lorlatinib is a brain-penetrant, third-generation ALK tyrosine kinase inhibitor indicated for the treatment of patients with *ALK*-positive metastatic NSCLC[1]
- Approval of lorlatinib in first line was based on the phase 3 CROWN study, which demonstrated significantly longer PFS and higher intracranial response with lorlatinib than crizotinib[2]
- After 5 years of follow-up, median PFS was not reached in the lorlatinib group, corresponding to the longest PFS for any single-agent molecular targeted treatment in advanced NSCLC[3]
- DepOR has been associated with survival outcomes in patients with metastatic NSCLC treated with targeted therapy[4]
- In this post hoc analysis of data from the CROWN study, the association between DepOR and PFS was assessed in the lorlatinib group

Methods

- The CROWN study (NCT03052608) is an ongoing, international, open-label, randomized, phase 3 trial comparing lorlatinib vs crizotinib in patients with previously untreated *ALK*-positive advanced NSCLC
 - Patients were randomized 1:1 to receive oral lorlatinib 100 mg once daily or crizotinib 250 mg twice daily
- DepOR is defined as the best percentage shrinkage in tumor size compared with baseline
- This post hoc analysis examined how DepOR is associated with demographics, baseline tumor characteristics, investigator-assessed PFS, and ctDNA–based biomarkers in the lorlatinib group
- Patients were evaluable for DepOR if they had target lesions at baseline and ≥1 adequate postbaseline assessment up to the time of progressive disease or new anticancer therapy
- Data cutoff for this analysis was October 31, 2023

Results

- In the lorlatinib group, 142 of 149 patients (95%) were evaluable for DepOR
 - The majority of patients (n=113 [80%]) experienced >50% shrinkage in target lesion (**Figure 1**)
 - 29 patients (20%) had 0% to 50% best target lesion shrinkage
 - 65 patients (46%) had >50% to 75% best target lesion shrinkage
 - 48 patients (34%) had >75% to 100% best target lesion shrinkage
- Demographics and baseline tumor characteristics were mostly similar across DepOR groups (**Table 1**); notably, a higher percentage of patients with baseline brain metastases was observed in the greater DepOR groups

Figure 1: Best target lesion shrinkage

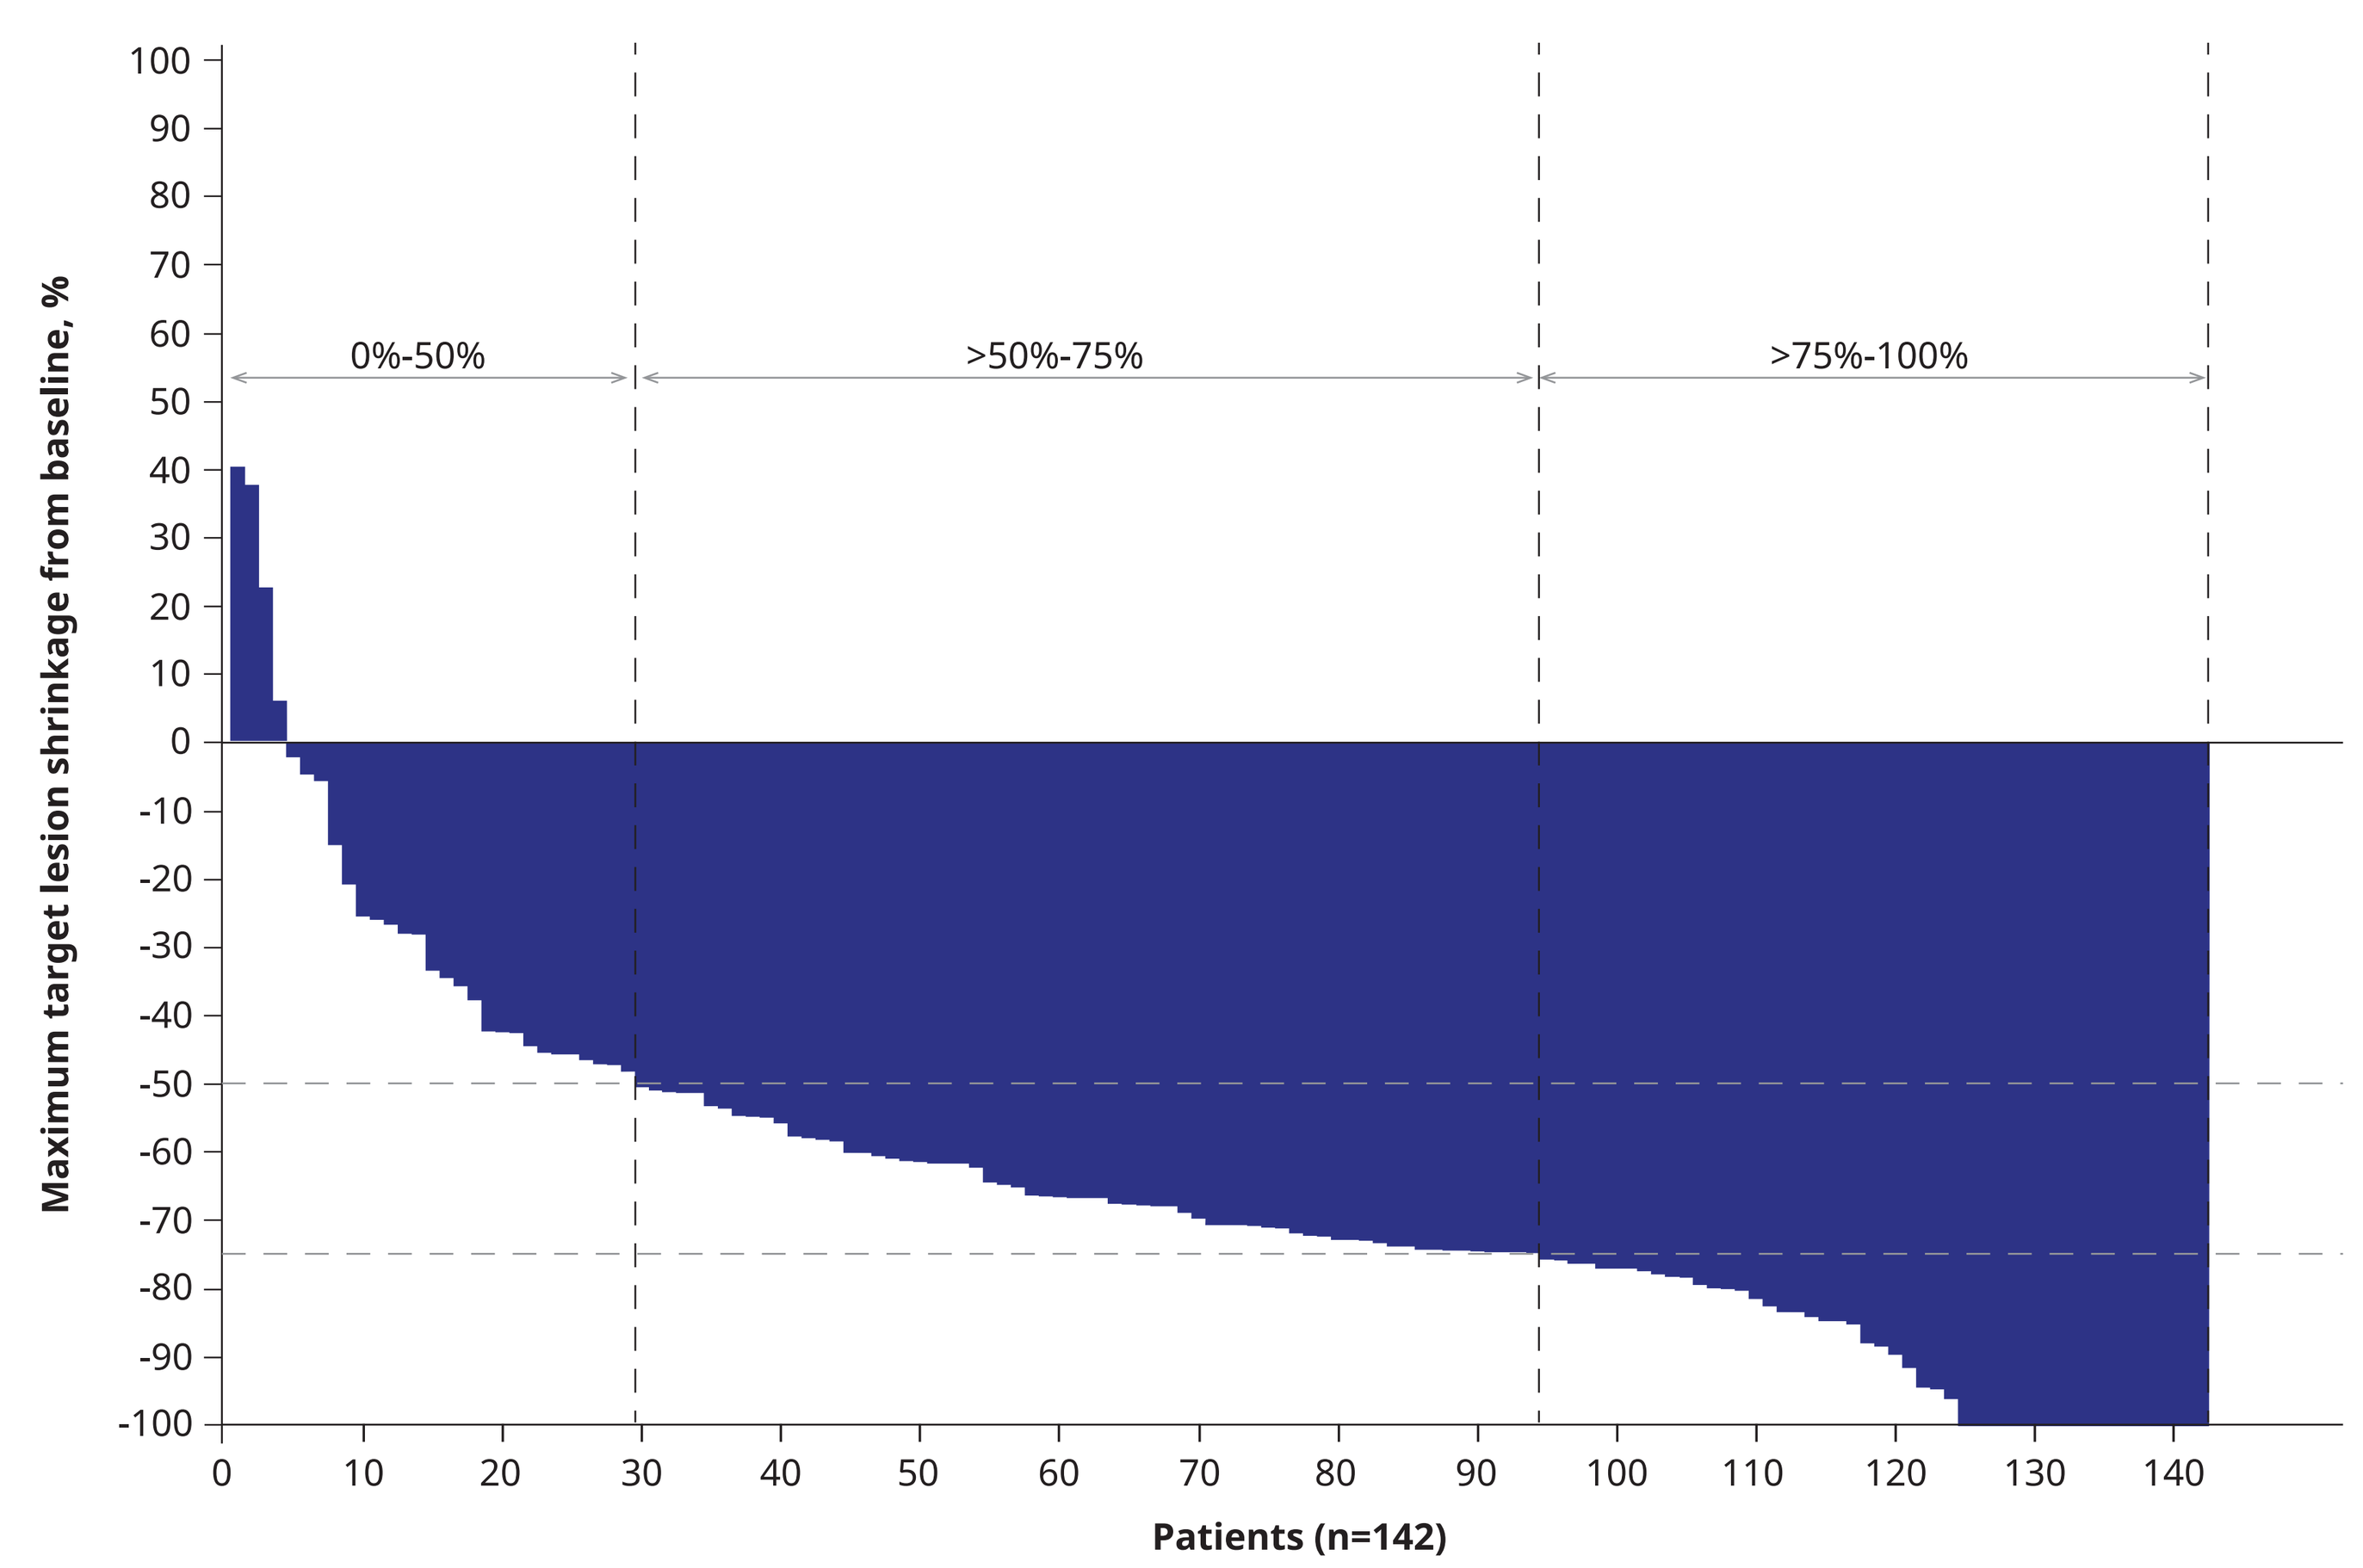
Maximum target lesion shrinkage from baseline, %
0%-50%
>50%-75%
>75%-100%
Patients (n=142)

Table 1: Demographics and baseline clinical characteristics in DepOR groups

	0%-50% (n=29)	>50%-75% (n=65)	>75%-100% (n=48)
Age, median (range), years	66 (51-70)	61 (52-70)	59 (48-67)
Sex, n (%)			
Female	15 (52)	41 (63)	25 (52)
Male	14 (48)	24 (37)	23 (48)
Race, n %			
Asian	12 (41)	32 (49)	18 (38)
White	17 (59)	29 (45)	23 (48)
Not reported	0	4 (6)	7 (15)
ECOG PS, n (%)			
0	16 (55)	28 (43)	21 (44)
1	13 (45)	36 (55)	26 (54)
2	0	1 (2)	1 (2)
Brain metastases at baseline, n (%)	5 (17)	15 (23)	15 (31)
Other involved tumor sites at baseline, n (%)			
Lung	26 (90)	58 (89)	40 (83)
Lymph nodes, regional and distant	21 (72)	54 (83)	37 (77)
Bone	9 (31)	18 (28)	19 (40)
Pleura	9 (31)	18 (28)	15 (31)
Liver	5 (17)	9 (14)	9 (19)
Adrenal glands	5 (17)	4 (6)	3 (6)
Other	6 (21)	10 (15)	7 (15)

DepOR, depth of response; ECOG PS, Eastern Cooperative Oncology Group performance status.

- Greater DepOR was associated with longer PFS (**Figure 2**)
 - In patients with 0% to 50% DepOR (n=29), median PFS was 12.7 months (95% CI, 7.2–not evaluable [NE])
 - In patients with >50% to 75% DepOR (n=65), median PFS was NE, with a hazard ratio (HR) of 0.39 (95% CI, 0.21–0.73) vs the 0%-50% group
 - In patients with >75% to 100% DepOR (n=48), median PFS was NE, with an HR of 0.25 (95% CI, 0.12–0.53) vs the 0%-50% group
- Of all plasma samples collected at screening (n=128), *EML4::ALK* long variant 1/2 was detected in 21%, and *EML4::ALK* short variant 3 was detected in 13%; *ALK* fusion was not detected in 25% of samples, and ctDNA was not detected in 24% (**Figure 3**)
- In the DepOR groups 0% to 50% (n=27), >50% to 75% (n=56), and >75% to 100% (n=45):
 - *EML4::ALK* variant 1/2 was detected in 15%, 27%, and 18% of samples, respectively
 - *EML4::ALK* variant 3 was detected in 7%, 14%, and 13% of samples, respectively
 - *ALK* fusion was not detected in 37%, 14%, and 31% of samples, respectively
 - ctDNA was not detected in 26%, 25%, and 22% of samples, respectively
- ctDNA dynamics at screening and cycle 2 day 1 (C2D1 or week 4) in DepOR groups 0% to 50% (n=22), >50% to 75% (n=53), and >75% to 100% (n=41) showed no association between DepOR and ctDNA clearance status (**Figure 4**)
 - ctDNA was not detected at either screening or C2D1 in 23%, 17%, and 15% of samples, respectively
 - ctDNA was detected at screening and not at C2D1 in 14%, 28%, and 32% of samples, respectively
 - ctDNA was not detected at screening and detected at C2D1 in 5%, 9%, and 10% of samples, respectively
 - ctDNA was detected at both screening and C2D1 in 59%, 45%, and 44% of samples, respectively

Figure 2: PFS by DepOR groups

	0%-50%	>50%-75%	>75%-100%
Events, n (%)	17 (59)	24 (37)	11 (23)
PFS, median (95% CI), months	12.7 (7.2–NE)	NE (60.0–NE)	NE (NE–NE)
Hazard ratio (95% CI)[a]		0.39 (0.21–0.73)	0.25 (0.12–0.53)

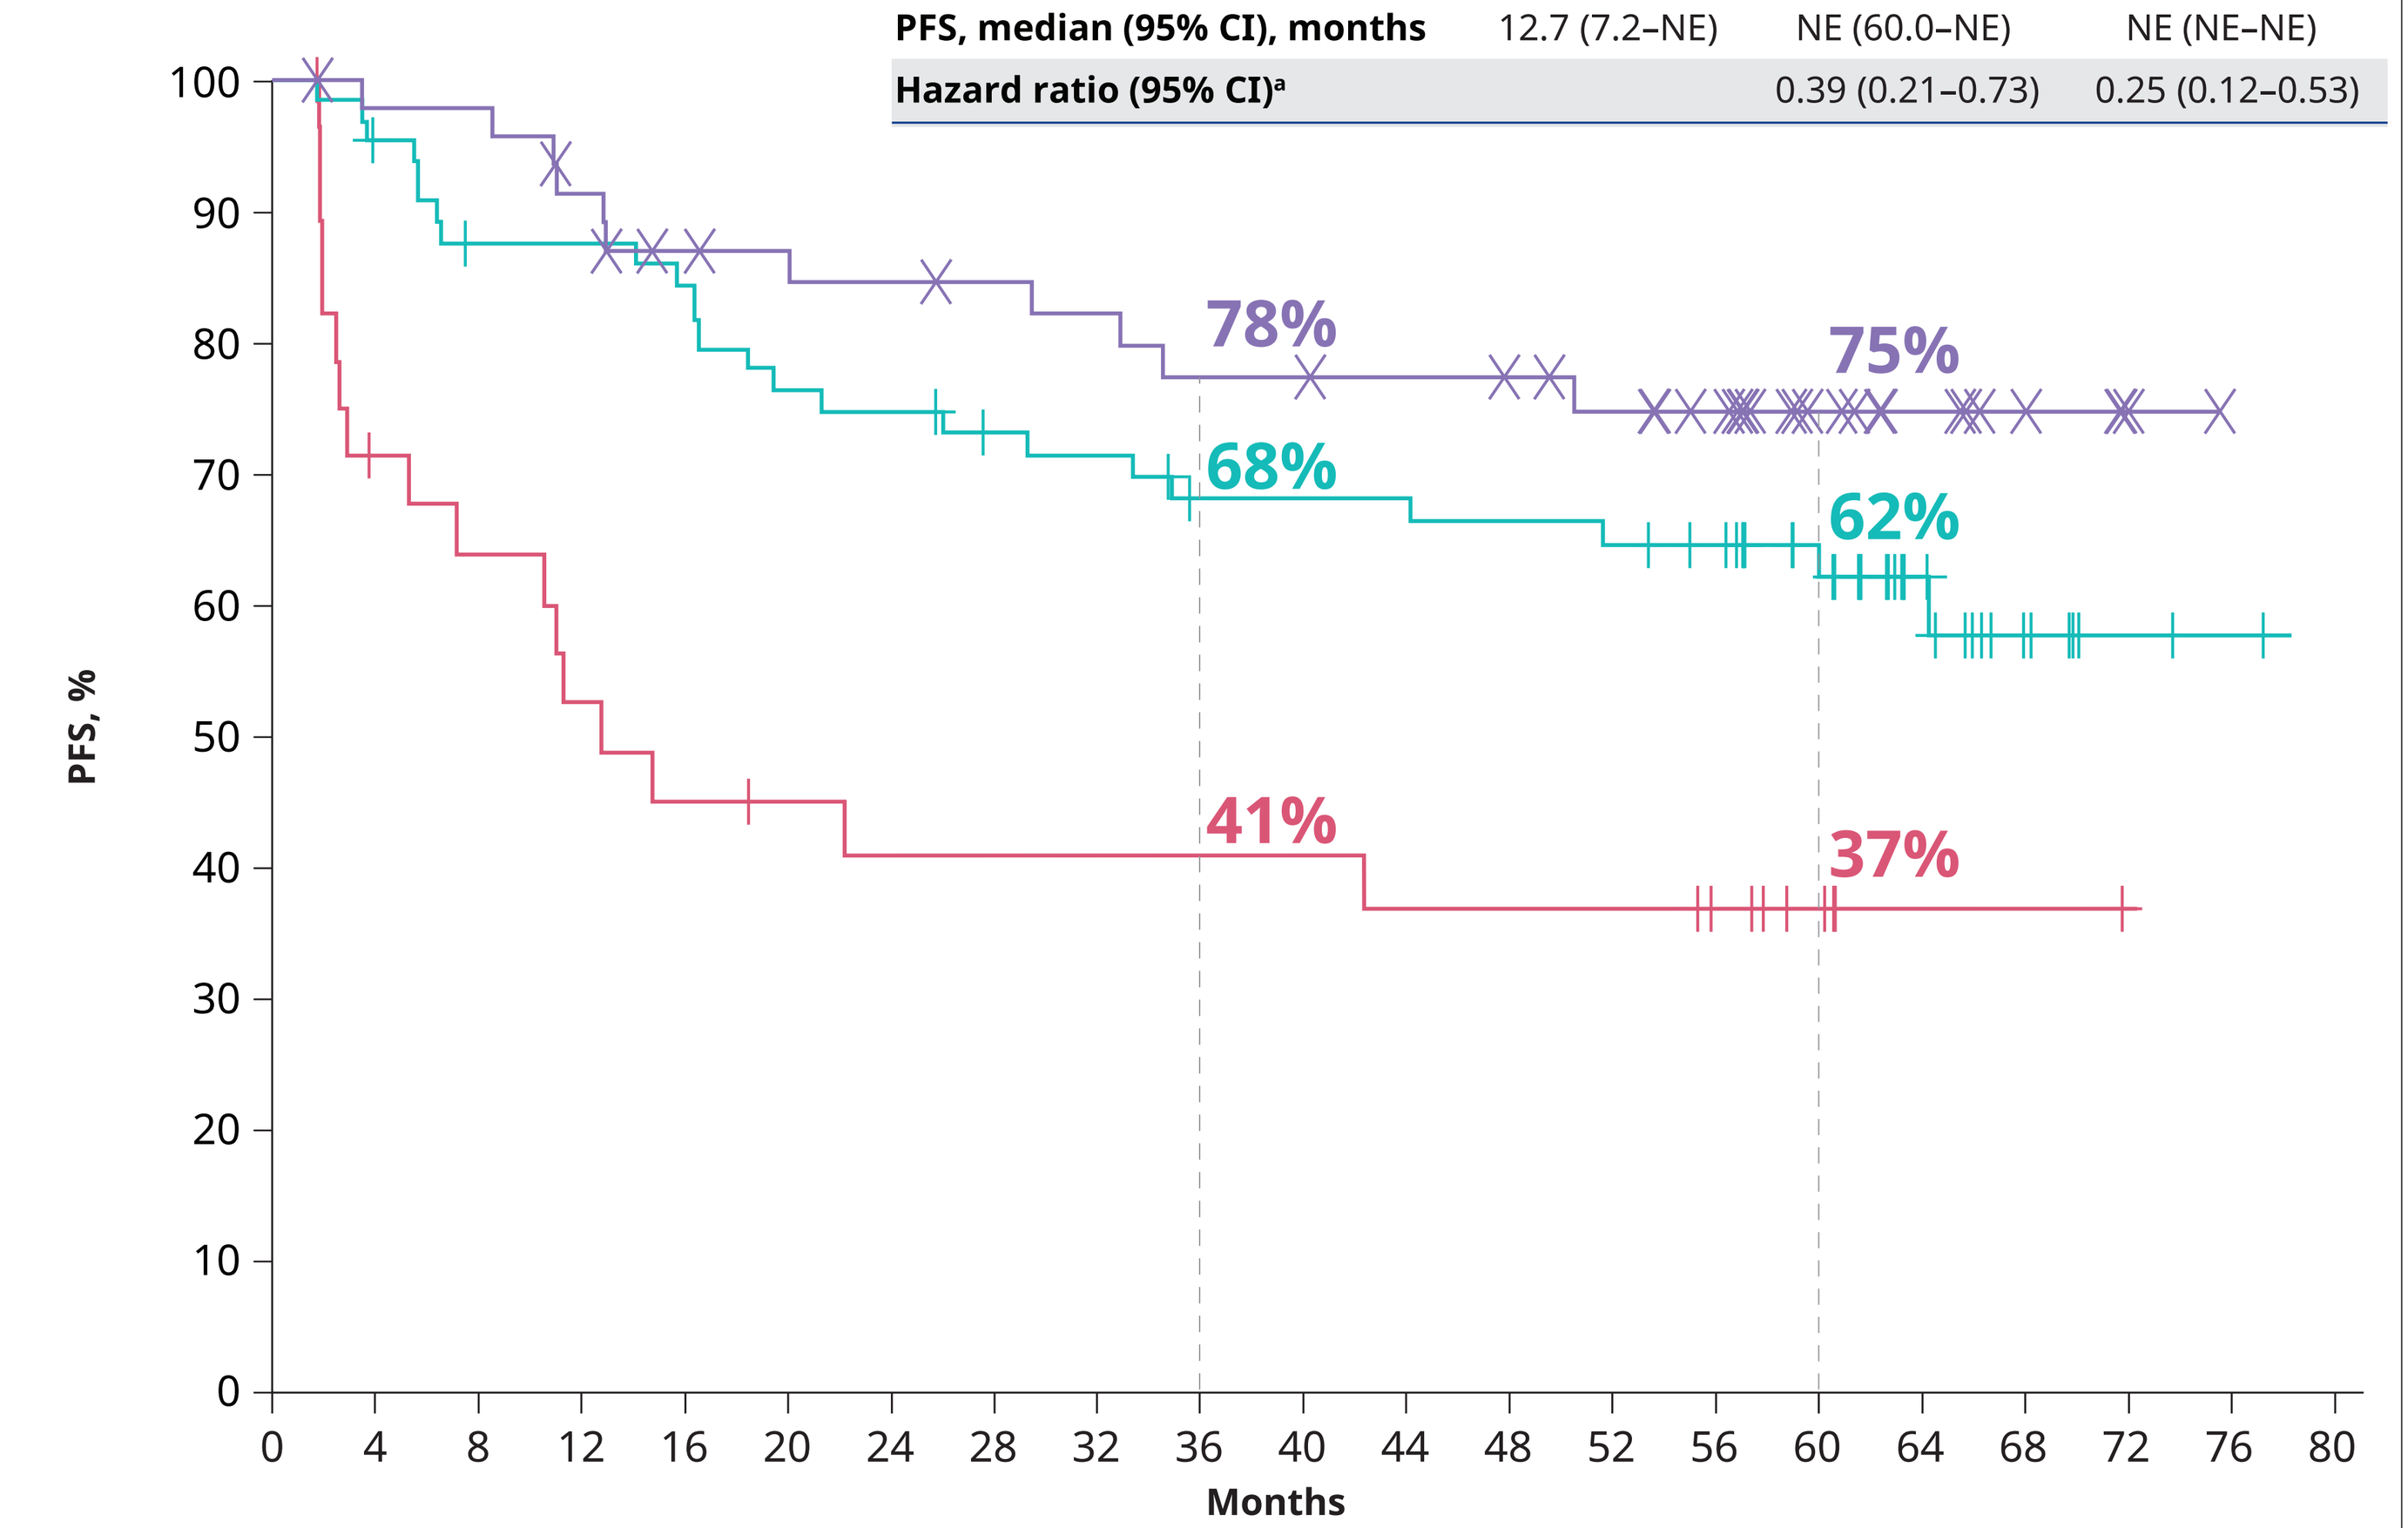
PFS, %
Months
78%
75%
68%
62%
41%
37%

No. at risk	0	4	8	12	16	20	24	28	32	36	40	44	48	52	56	60	64	68	72	76	80
0%-50%	29	19	17	14	12	11	10	10	10	10	10	9	9	9	7	4	1	1	0	0	0
>50%-75%	65	61	55	55	53	48	47	44	43	39	39	39	38	37	35	26	15	6	2	1	0
>75%-100%	48	46	46	42	38	37	36	35	34	32	32	31	30	28	25	15	10	7	2	0	0

DepOR, depth of response; NE, not evaluable; PFS, progression-free survival.
[a]Unstratified analysis comparison vs 0%-50% group.

Figure 3: *ALK* fusion variants at screening by DepOR groups

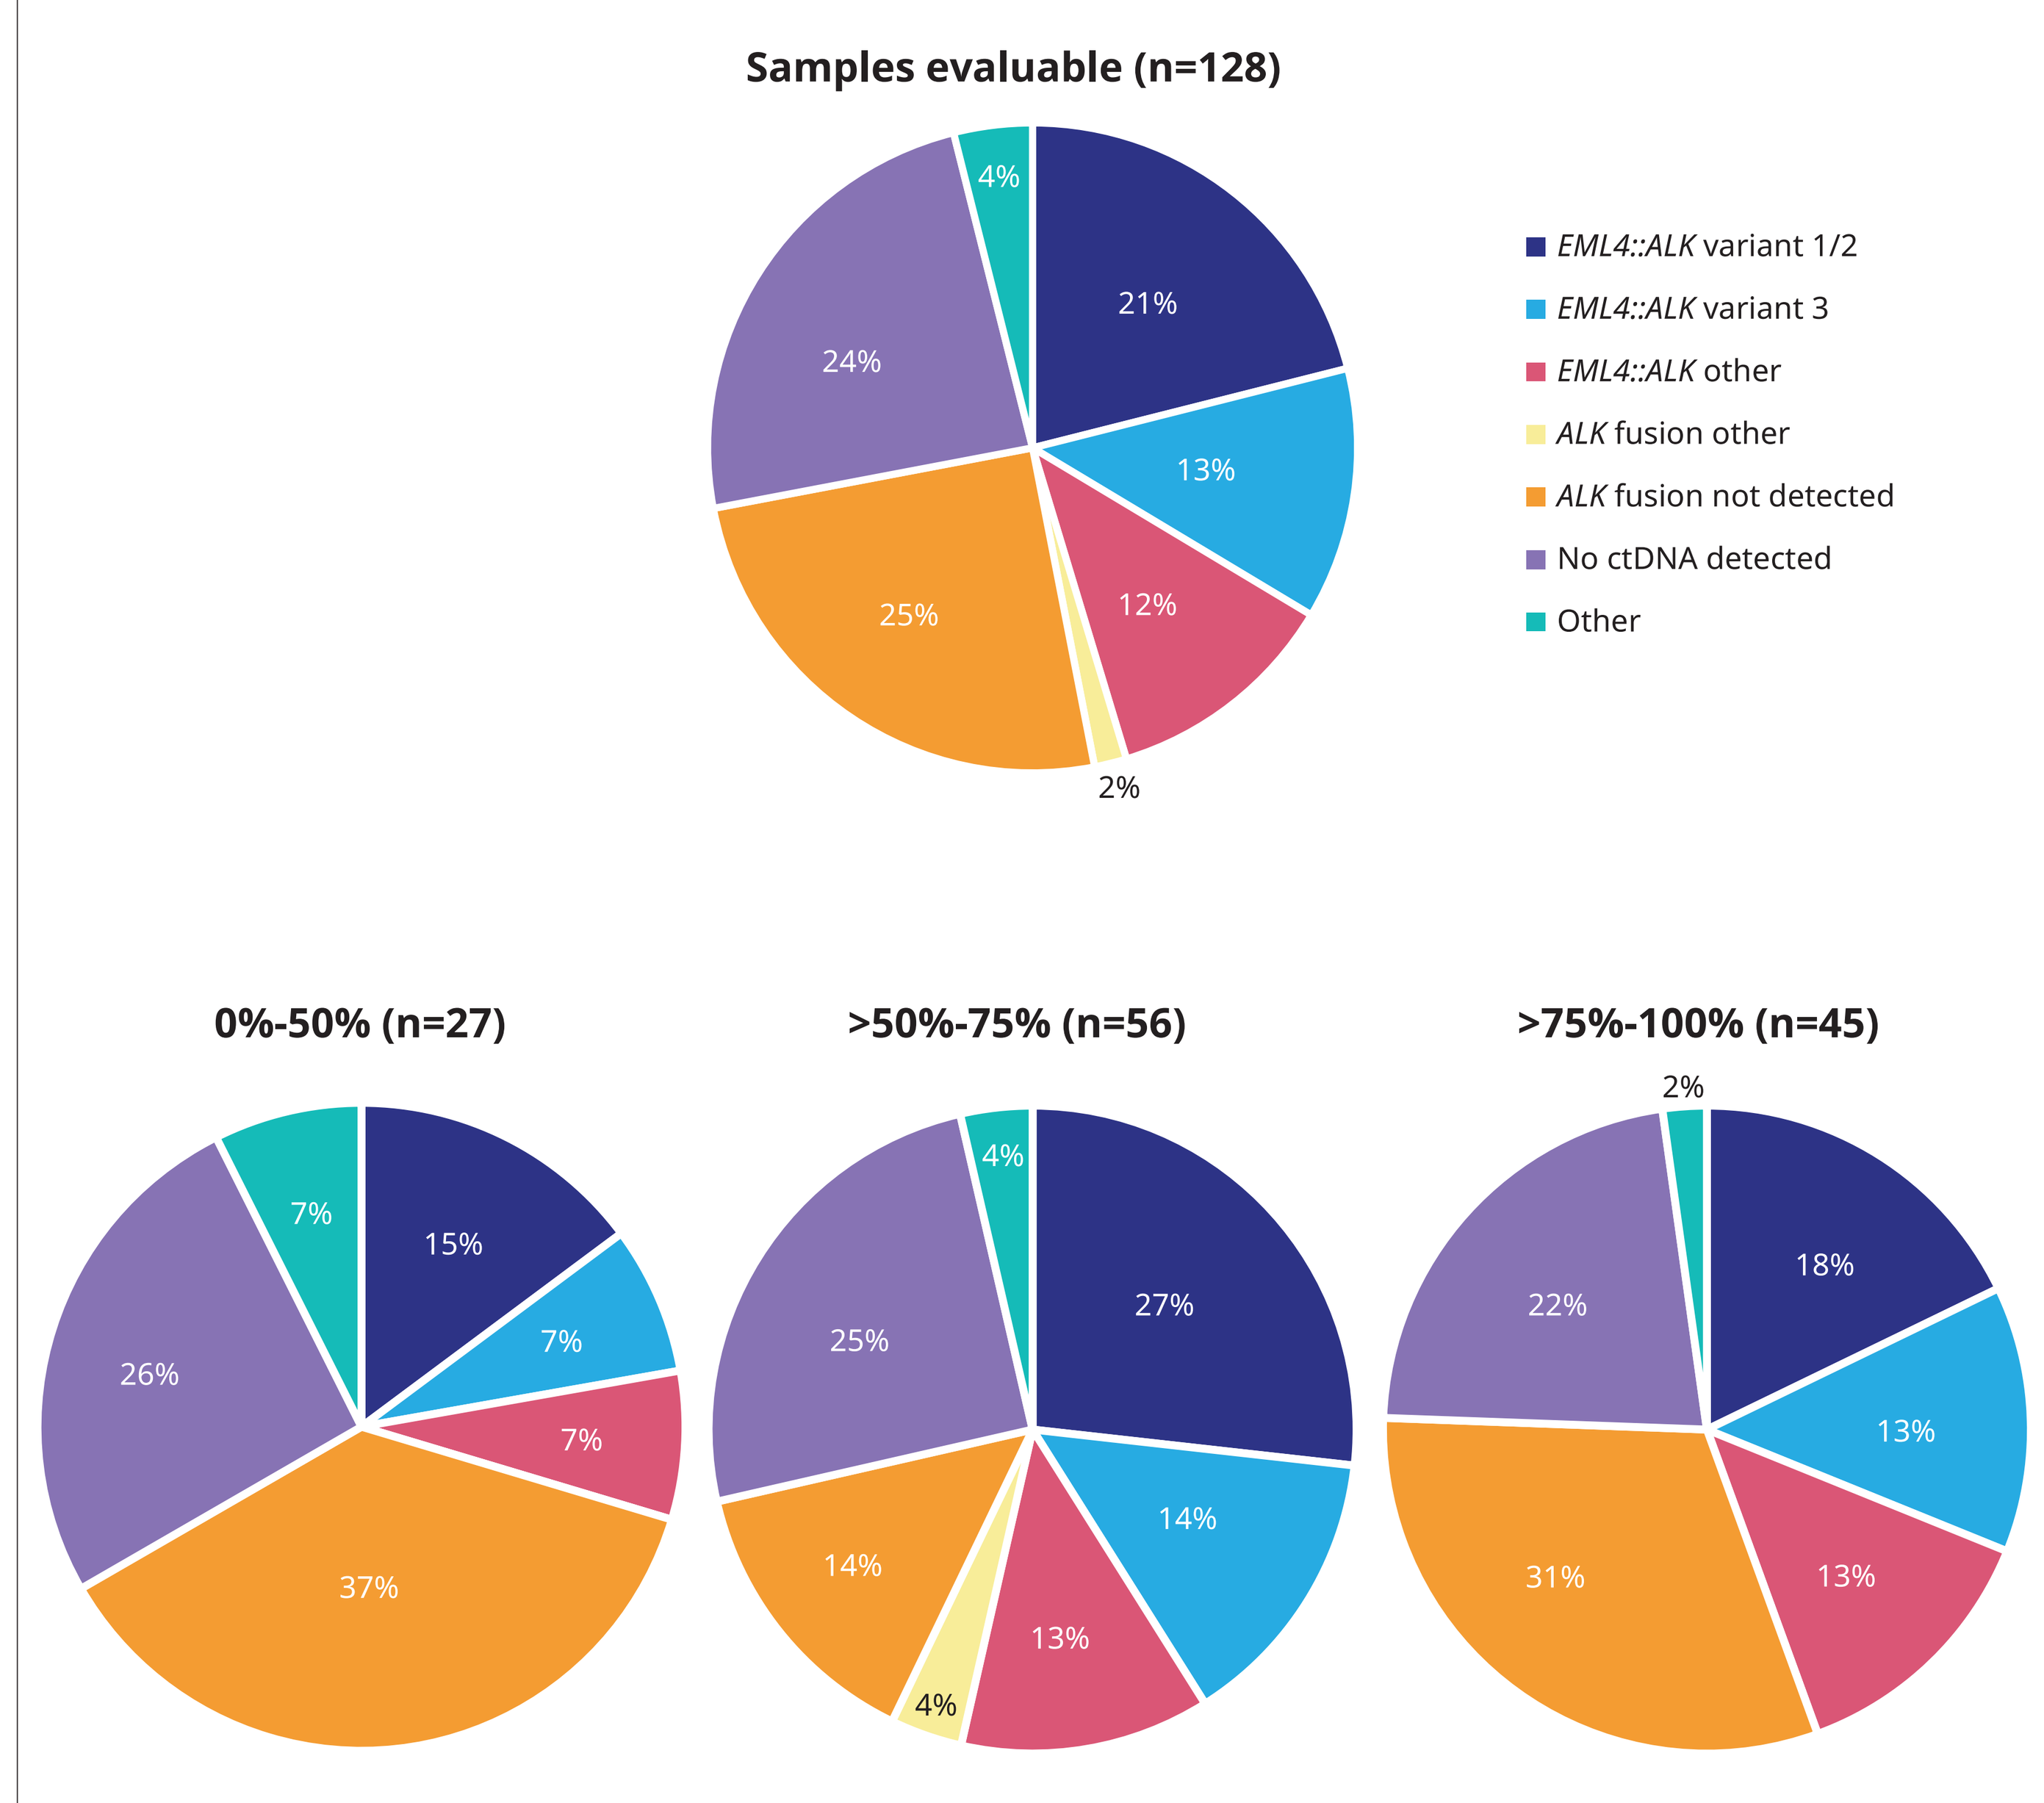
Samples evaluable (n=128)
21%
13%
12%
2%
25%
24%
4%
EML4::ALK variant 1/2
EML4::ALK variant 3
EML4::ALK other
ALK fusion other
ALK fusion not detected
No ctDNA detected
Other
0%-50% (n=27)
15%
7%
7%
37%
26%
7%
>50%-75% (n=56)
27%
14%
13%
4%
14%
25%
4%
>75%-100% (n=45)
18%
13%
13%
31%
22%
2%

ctDNA, circulating tumor DNA; DepOR, depth of response.

Figure 4: ctDNA dynamics at week 4 (C2D1) by DepOR groups

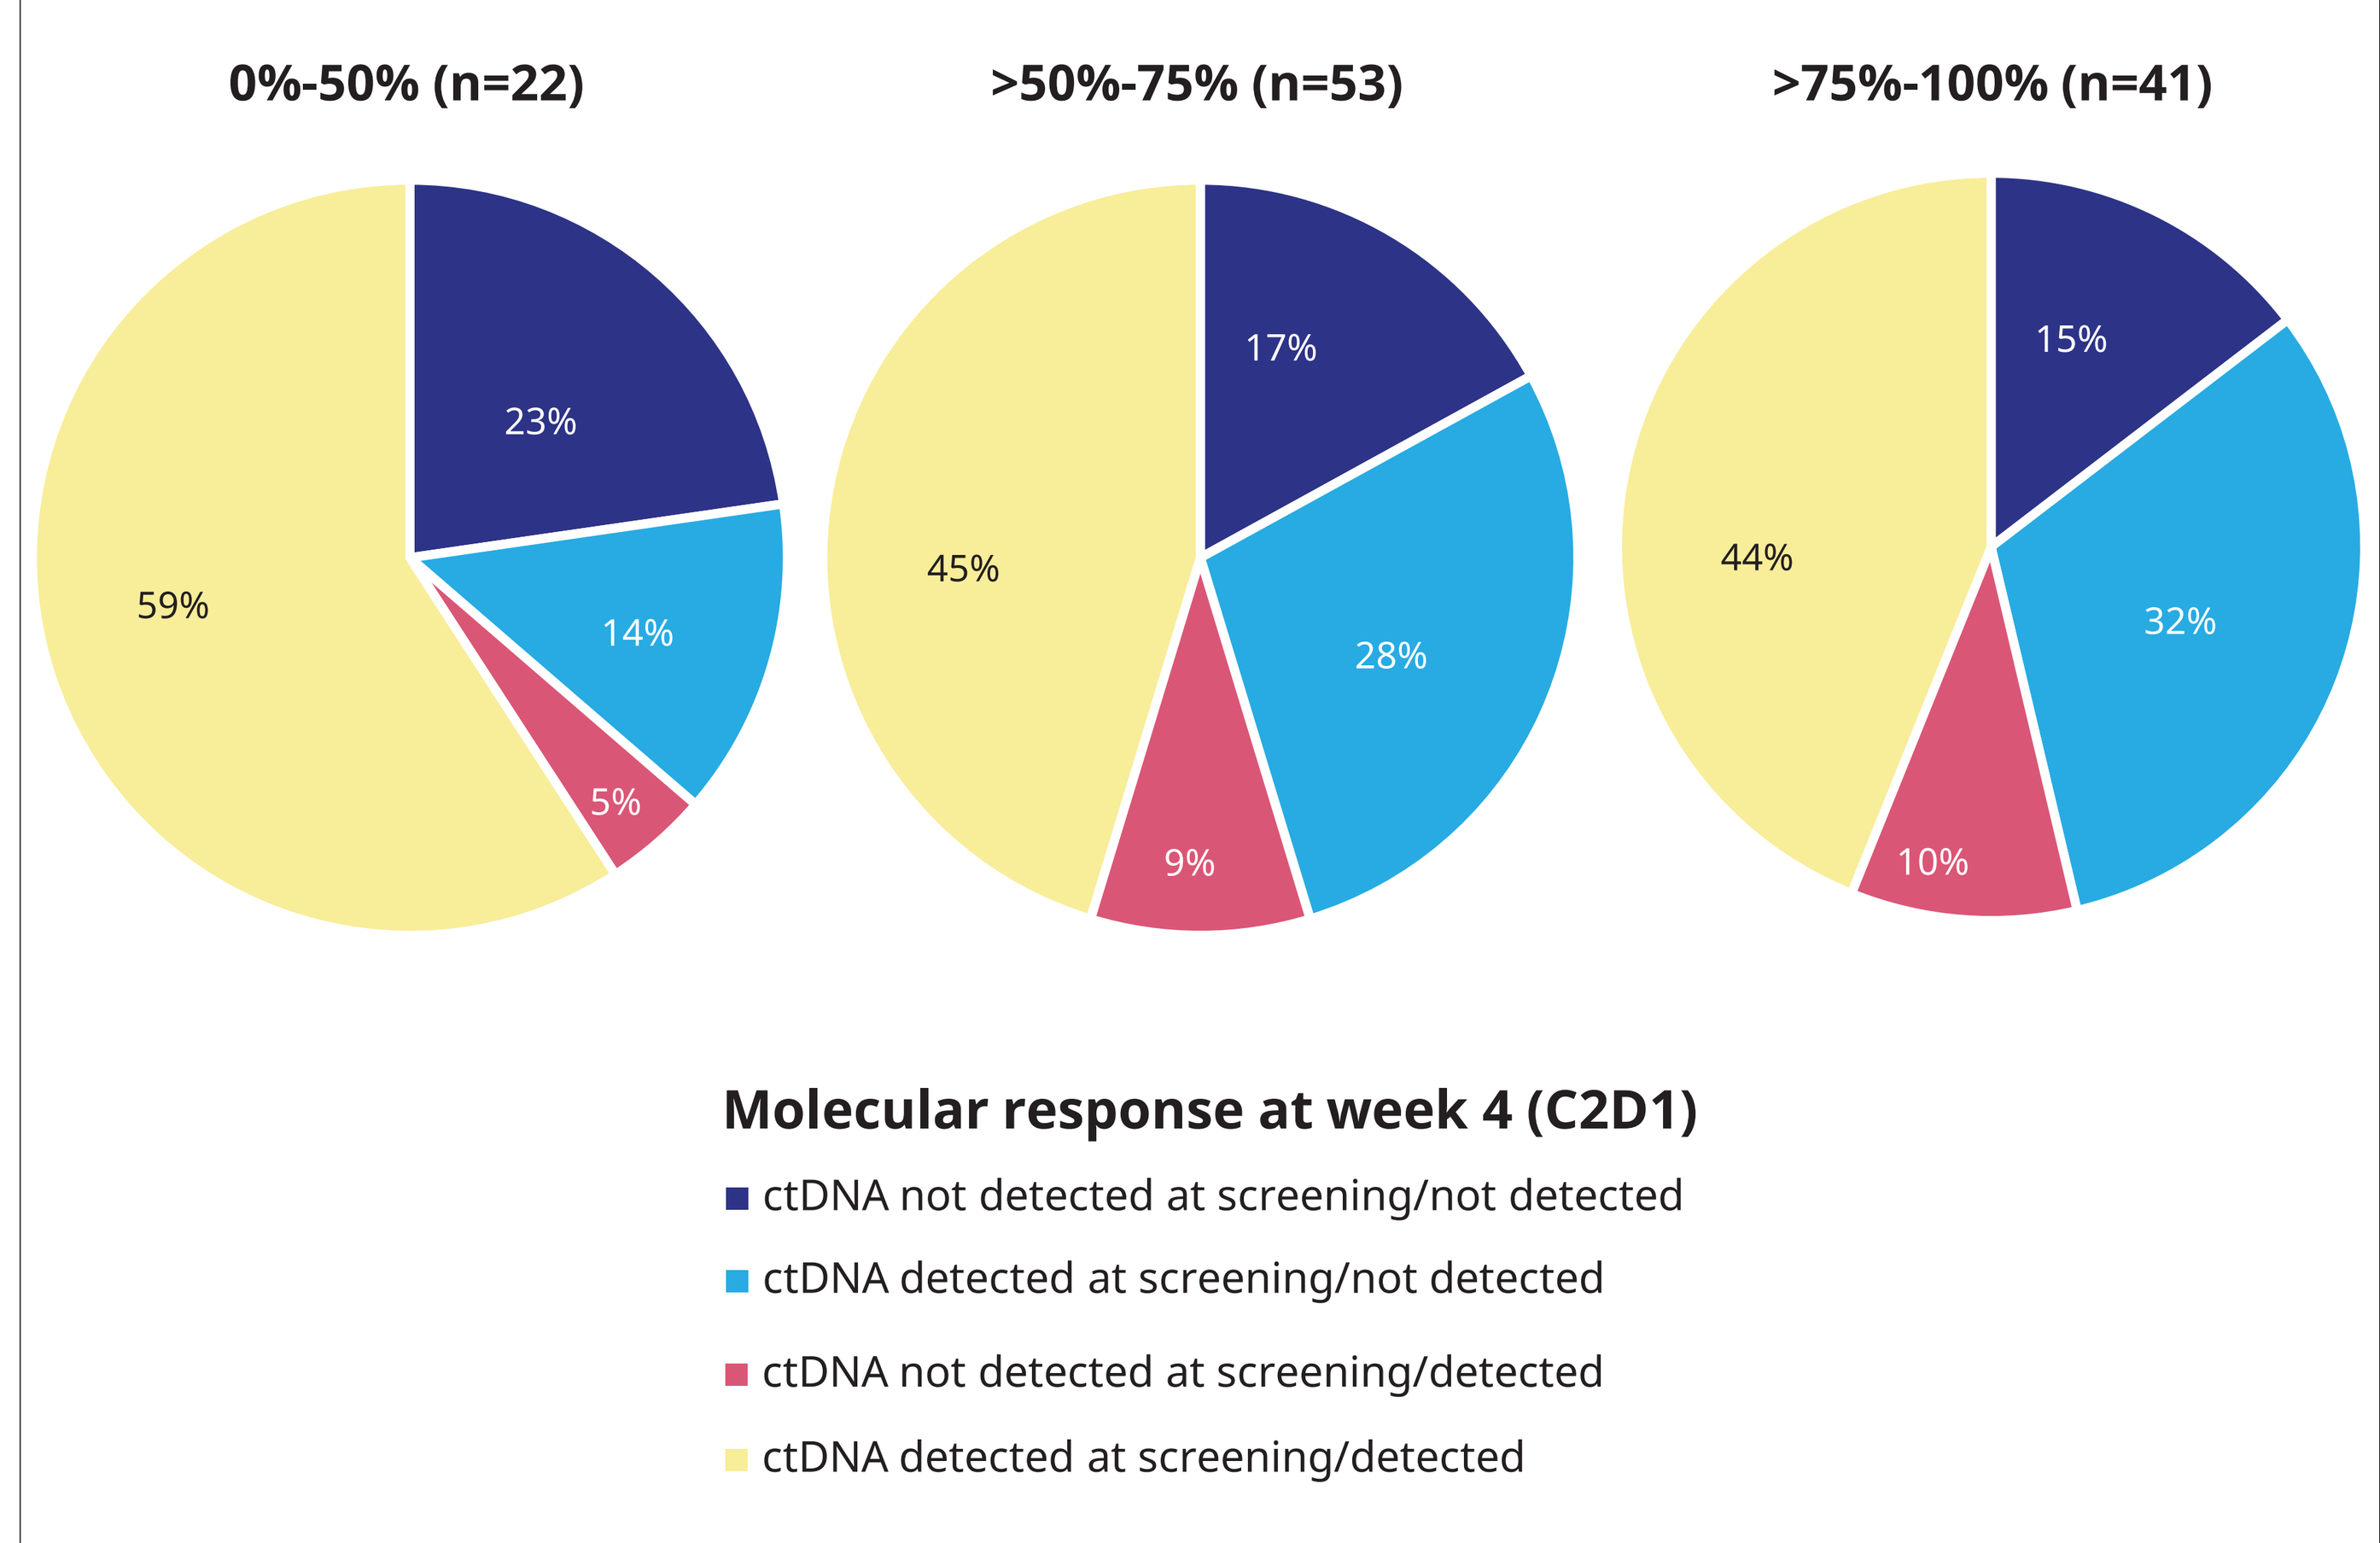
0%-50% (n=22)
23%
14%
5%
59%
>50%-75% (n=53)
17%
28%
9%
45%
>75%-100% (n=41)
15%
32%
10%
44%
Molecular response at week 4 (C2D1)
ctDNA not detected at screening/not detected
ctDNA detected at screening/not detected
ctDNA not detected at screening/detected
ctDNA detected at screening/detected

C2D1, cycle 2 day 1; ctDNA, circulating tumor DNA; DepOR, depth of response.