P-2017

Patient-Reported Outcomes of Nirmatrelvir/Ritonavir Treatment for High-Risk, Nonhospitalized Adults With Symptomatic COVID-19

Ashley S. Cha-Silva,[1] Henriette Coetzer,[2] Kelly Gebo,[3] Jinma Ren,[4] Amie Scott,[5] Joseph C. Cappelleri,[6] Wajeeha Ansari,[1] Heidi Leister-Tebbe[7]

[1]Global Biopharmaceuticals Group, Pfizer Inc, New York, NY, USA; [2]Blue Health Intelligence® (BHI®), Chicago, IL, USA; [3]Johns Hopkins University School of Medicine, Baltimore, MD, USA; [4]Global Biometrics and Data Management, Pfizer Inc, Collegeville, PA, USA; [5]Global Real World Evidence, Pfizer Inc, New York, NY, USA; [6]Global Biometrics and Data Management, Pfizer Inc, Groton, CT, USA; [7]Pfizer Research and Development, Pfizer Inc, Collegeville, PA, USA

BACKGROUND AND OBJECTIVES

- Although >97% of US adults have antibodies against SARS-CoV-2 through vaccination and/or infection, new variants continue to evade the immune response, driving disease and mortality and straining global healthcare systems.[1-3]
- Beyond its serious effects on physical health, COVID-19 substantially impacts daily activities and quality of life for many patients.[4-5]
- Nirmatrelvir/ritonavir (NMV/r; Paxlovid, Pfizer Inc) is approved in the United States and >70 other countries worldwide for the treatment of mild-to-moderate COVID-19 in vaccinated and unvaccinated adults who are at high risk of progression to severe disease.[6,7]
- NMV/r is recommended by the US National Institutes of Health and the World Health Organization as the first line of treatment for COVID-19 in this high-risk population.[8,9]
- In the phase 2/3 EPIC-HR (Evaluation of Protease Inhibition for COVID-19 in High-Risk Patients) study, NMV/r demonstrated an 86% relative risk reduction vs placebo against COVID-19–related hospitalization or all-cause mortality in high-risk adults with mild-to-moderate COVID-19.[7,10]
 - Patient-reported outcomes (PROs) were also collected in EPIC-HR to determine how NMV/r impacted patient assessment of their symptom burden, work productivity, daily activities, and overall health-related quality of life (HRQoL).
- Here, we report PROs from EPIC-HR using results from the Global Impression Questions (GIQ), Work Productivity and Activity Impairment (WPAI-COVID-19), and EuroQol Quality-of-Life 5-Dimension 5-Level Scale (EQ-5D-5L) questionnaires.

METHODS

- In EPIC-HR, nonhospitalized adults at high risk of progression to severe COVID-19 were randomized 1:1 within 5 days of symptom onset to receive either NMV/r or placebo twice daily for 5 days.
- Patients were asked to complete the GIQ at baseline and daily for 28 days using an electronic diary and to complete the WPAI-COVID-19 (adapted from the General Health WPAI)[11] and EQ-5D-5L[12] at specified visits through Week 24.
- PRO analyses included randomized patients who received ≥1 dose of study intervention and had ≥1 postbaseline visit through Day 28 (modified intent-to-treat 2 [mITT2] population).
 - Rollout of WPAI-COVID-19 and EQ-5D-5L questionnaires occurred after study initiation, limiting response availability.
- The GIQ[13] consisted of 3 questions regarding patients' return to usual health, return to usual activities, and COVID-19 symptom severity.
 - The time to return to usual health or activities and time to resolution or alleviation of symptoms were calculated as the first event date minus the number of days from receipt of the first dose, plus 1 day.
 - Sustained resolution of symptoms was defined as the first of 4 consecutive days with no reported overall symptoms.
 - Sustained alleviation of symptoms was defined as the first of either 1) 4 consecutive days with mild or absent symptoms for symptoms reported as moderate or severe at baseline or 2) 4 consecutive days with absent symptoms for symptoms reported as mild at baseline.
- GIQ results were analyzed using the Kaplan-Meier method with a log-rank test and a Cox proportional hazard model to calculate hazard ratios (HRs) with 95% CIs.[14]
- In the Cox proportional hazard analysis, treatment group (NMV/r or placebo) and geographic region were used as independent variables and controlled covariates included symptom onset duration (≤3 or >3 days), COVID-19 monoclonal antibody treatment (yes or no), baseline SARS-CoV-2 serology status (positive or negative), and baseline viral load (<4 or ≥4 $\log_{10}$ copies/mL).

RESULTS

- Out of 2246 randomized patients, 2091 were included in the mITT2 population (NMV/r, n=1038; placebo, n=1053; **Figure 1**).

Figure 1. CONSORT Diagram Detailing Enrollment and Randomization

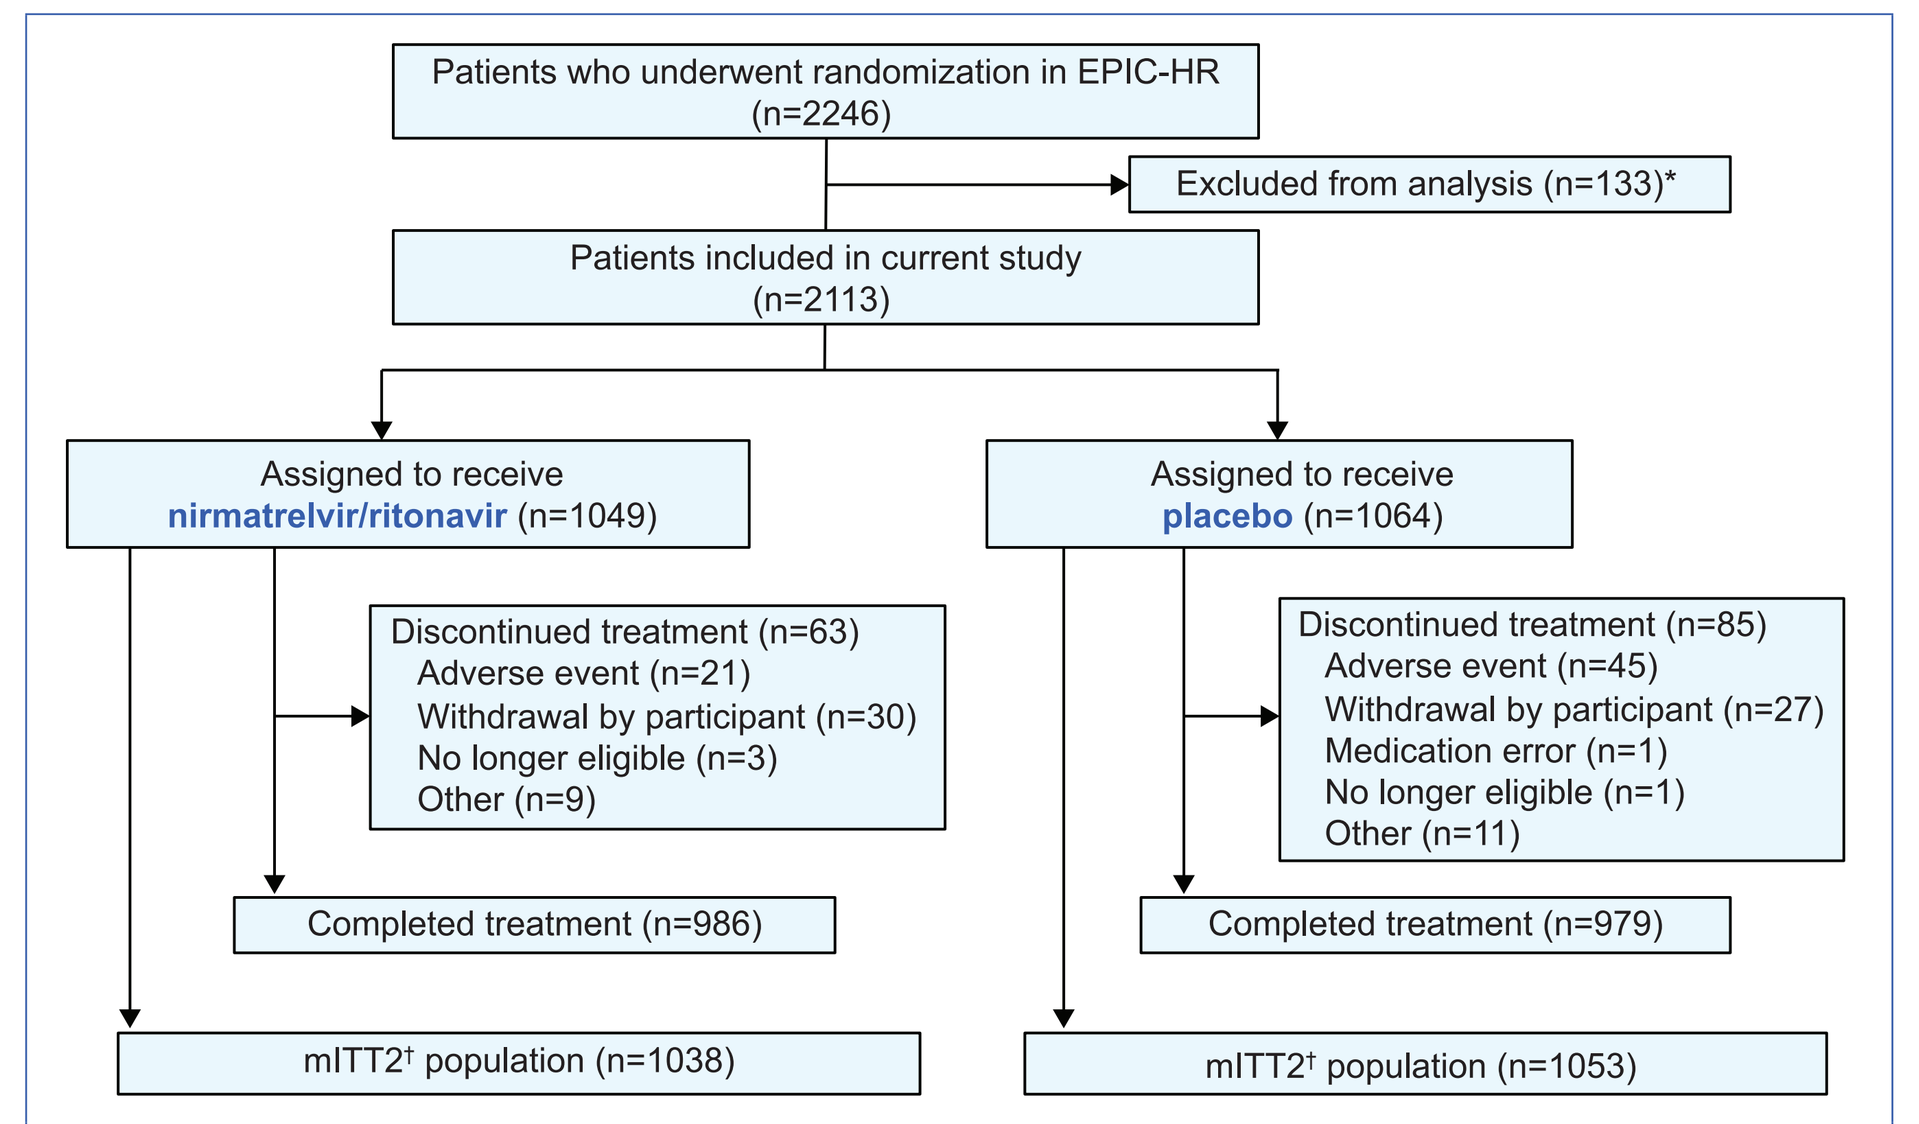
Patients who underwent randomization in EPIC-HR (n=2246)
Excluded from analysis (n=133)*
Patients included in current study (n=2113)
Assigned to receive nirmatrelvir/ritonavir (n=1049)
Assigned to receive placebo (n=1064)
Discontinued treatment (n=63)
Adverse event (n=21)
Withdrawal by participant (n=30)
No longer eligible (n=3)
Other (n=9)
Discontinued treatment (n=85)
Adverse event (n=45)
Withdrawal by participant (n=27)
Medication error (n=1)
No longer eligible (n=1)
Other (n=11)
Completed treatment (n=986)
Completed treatment (n=979)
mITT2† population (n=1038)
mITT2† population (n=1053)

mITT2=modified intent-to-treat 2.
*Data were excluded from 2 sites from the full analysis set due to data quality issues. †mITT2 population includes all patients who received ≥1 dose of the study intervention and had ≥1 postbaseline visit through Day 28.

- Demographic and baseline clinical characteristics were similar between treatment groups **(Table 1)**.

Table 1. Patient Demographic and Baseline Clinical Characteristics of the mITT2 Population

Characteristic	Nirmatrelvir/ Ritonavir 300 mg/100 mg (n=1038)	Placebo (n=1053)	Total (N=2091)
Age, median (range), years	44.0 (18.0–86.0)	46.0 (18.0–88.0)	45.0 (18.0–88.0)
Male, n (%)	516 (49.7)	538 (51.1)	1054 (50.4)
Race, n (%)			
White	728 (70.1)	756 (71.8)	1484 (71.0)
Black or African American	52 (5.0)	35 (3.3)	87 (4.2)
Asian	153 (14.7)	156 (14.8)	309 (14.8)
American Indian or Alaska Native	95 (9.2)	94 (8.9)	189 (9.0)
Not reported	8 (0.8)	9 (0.9)	17 (0.8)
BMI, mean (SD), kg/m^2	29.0 (5.4)	29.2 (5.7)	29.1 (5.5)
Geographic region, n (%)			
United States	387 (37.3)	399 (37.9)	786 (37.6)
Europe	330 (31.8)	333 (31.6)	663 (31.7)
India	94 (9.1)	97 (9.2)	191 (9.1)
Rest of world	227 (21.9)	224 (21.3)	451 (21.6)
Duration since first diagnosis, mean (SD), days	1.2 (1.3)	1.3 (1.2)	1.2 (1.2)
Duration since first symptom, mean (SD), days	2.9 (1.1)	3.0 (1.1)	2.9 (1.1)
Number of risk factors of interest, n (%)			
1	434 (41.8)	406 (38.6)	840 (40.2)
2	364 (35.1)	381 (36.2)	745 (35.6)
3	161 (15.5)	179 (17.0)	340 (16.3)
≥4	78 (7.5)	87 (8.3)	165 (7.9)
SARS-CoV-2 serology status, n (%)			
Negative	504 (48.6)	526 (50.0)	1030 (49.3)
Positive	521 (50.2)	514 (48.8)	1035 (49.5)
Unknown	13 (1.3)	13 (1.2)	26 (1.2)
Viral load, mean (SD), $\log_{10}$ copies/mL	4.8 (2.9)	4.7 (2.9)	4.7 (2.9)

BMI=body mass index; mITT2=modified intent-to-treat 2; SD=standard deviation.

- Among patients who completed the GIQ on Day 28, the time to return to usual health and time to return to usual activities were significantly reduced for patients who received NMV/r compared with placebo **(Figure 2, Figure 3)**.
 - By Day 28, 81% of patients receiving NMV/r vs 76% of patients receiving placebo reported a return to usual health **(Figure 2)**.
 - Differences between the 2 groups were first apparent on Day 4.
 - Patients treated with NMV/r had a significant 3-day reduction in median time to return to usual health (10 [95% CI, 10–11] days) compared with placebo (13 [95% CI, 12–14] days; HR, 1.27 [95% CI, 1.2–1.4]; *P*<0.0001).
 - By Day 28, 88% of patients receiving NMV/r vs 83% of patients receiving placebo reported a return to usual activities **(Figure 3)**.
 - Patients treated with NMV/r had a significant 1-day reduction in median time to return to usual activities (11 [95% CI, 10–11] days) compared with placebo (12 [95% CI, 11–12] days; HR, 1.2 [95% CI, 1.1–1.4]; *P*<0.0001).

Figure 2. Percentage of Patients in Each Treatment Group in the mITT2 Population Reporting Return to Usual Health on Each Study Day Through Day 28 on the Global Impressions Questionnaire

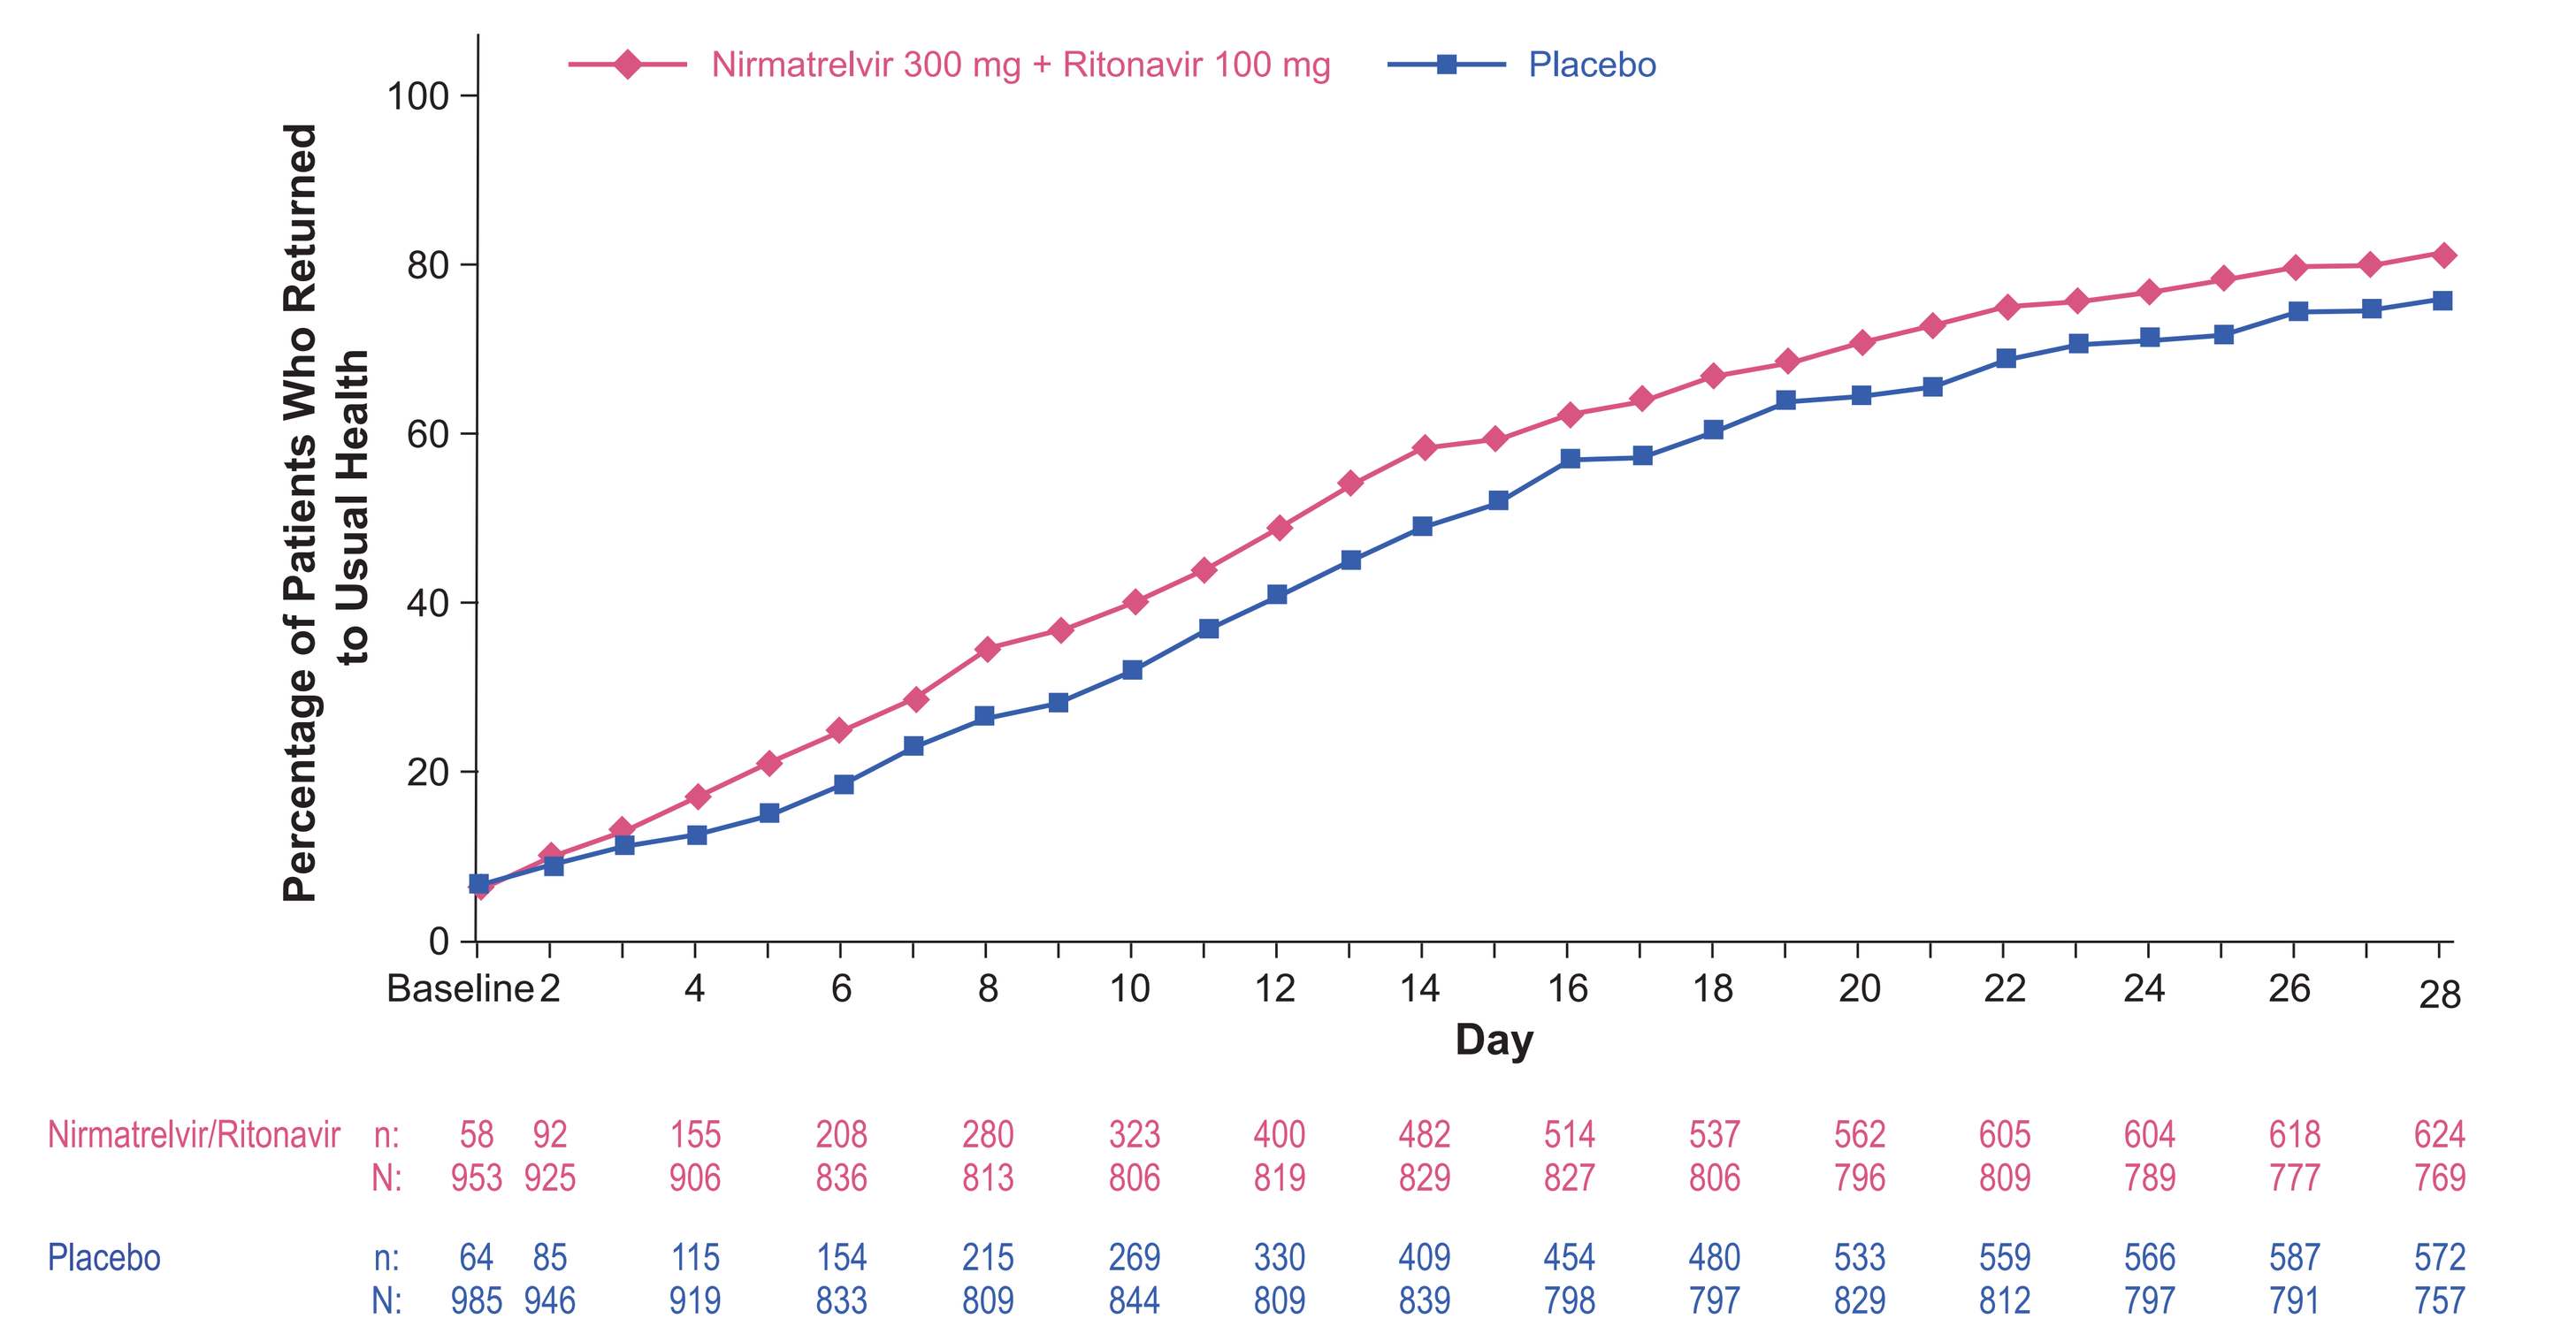
Nirmatrelvir 300 mg + Ritonavir 100 mg
Placebo
Percentage of Patients Who Returned to Usual Health
Day
Baseline 2 4 6 8 10 12 14 16 18 20 22 24 26 28
Nirmatrelvir/Ritonavir n: 58 92 155 208 280 323 400 482 514 537 562 605 604 618 624
N: 953 925 906 836 813 806 819 829 827 806 796 809 789 777 769
Placebo n: 64 85 115 154 215 269 330 409 454 480 533 559 566 587 572
N: 985 946 919 833 809 844 809 839 798 797 829 812 797 791 757

mITT2=modified intent-to-treat 2; n=number of patients with event; N=number of patients with nonmissing data.

Figure 3. Percentage of Patients in Each Treatment Group in the mITT2 Population Reporting Return to Usual Activities on Each Study Day Through Day 28 on the Global Impressions Questionnaire

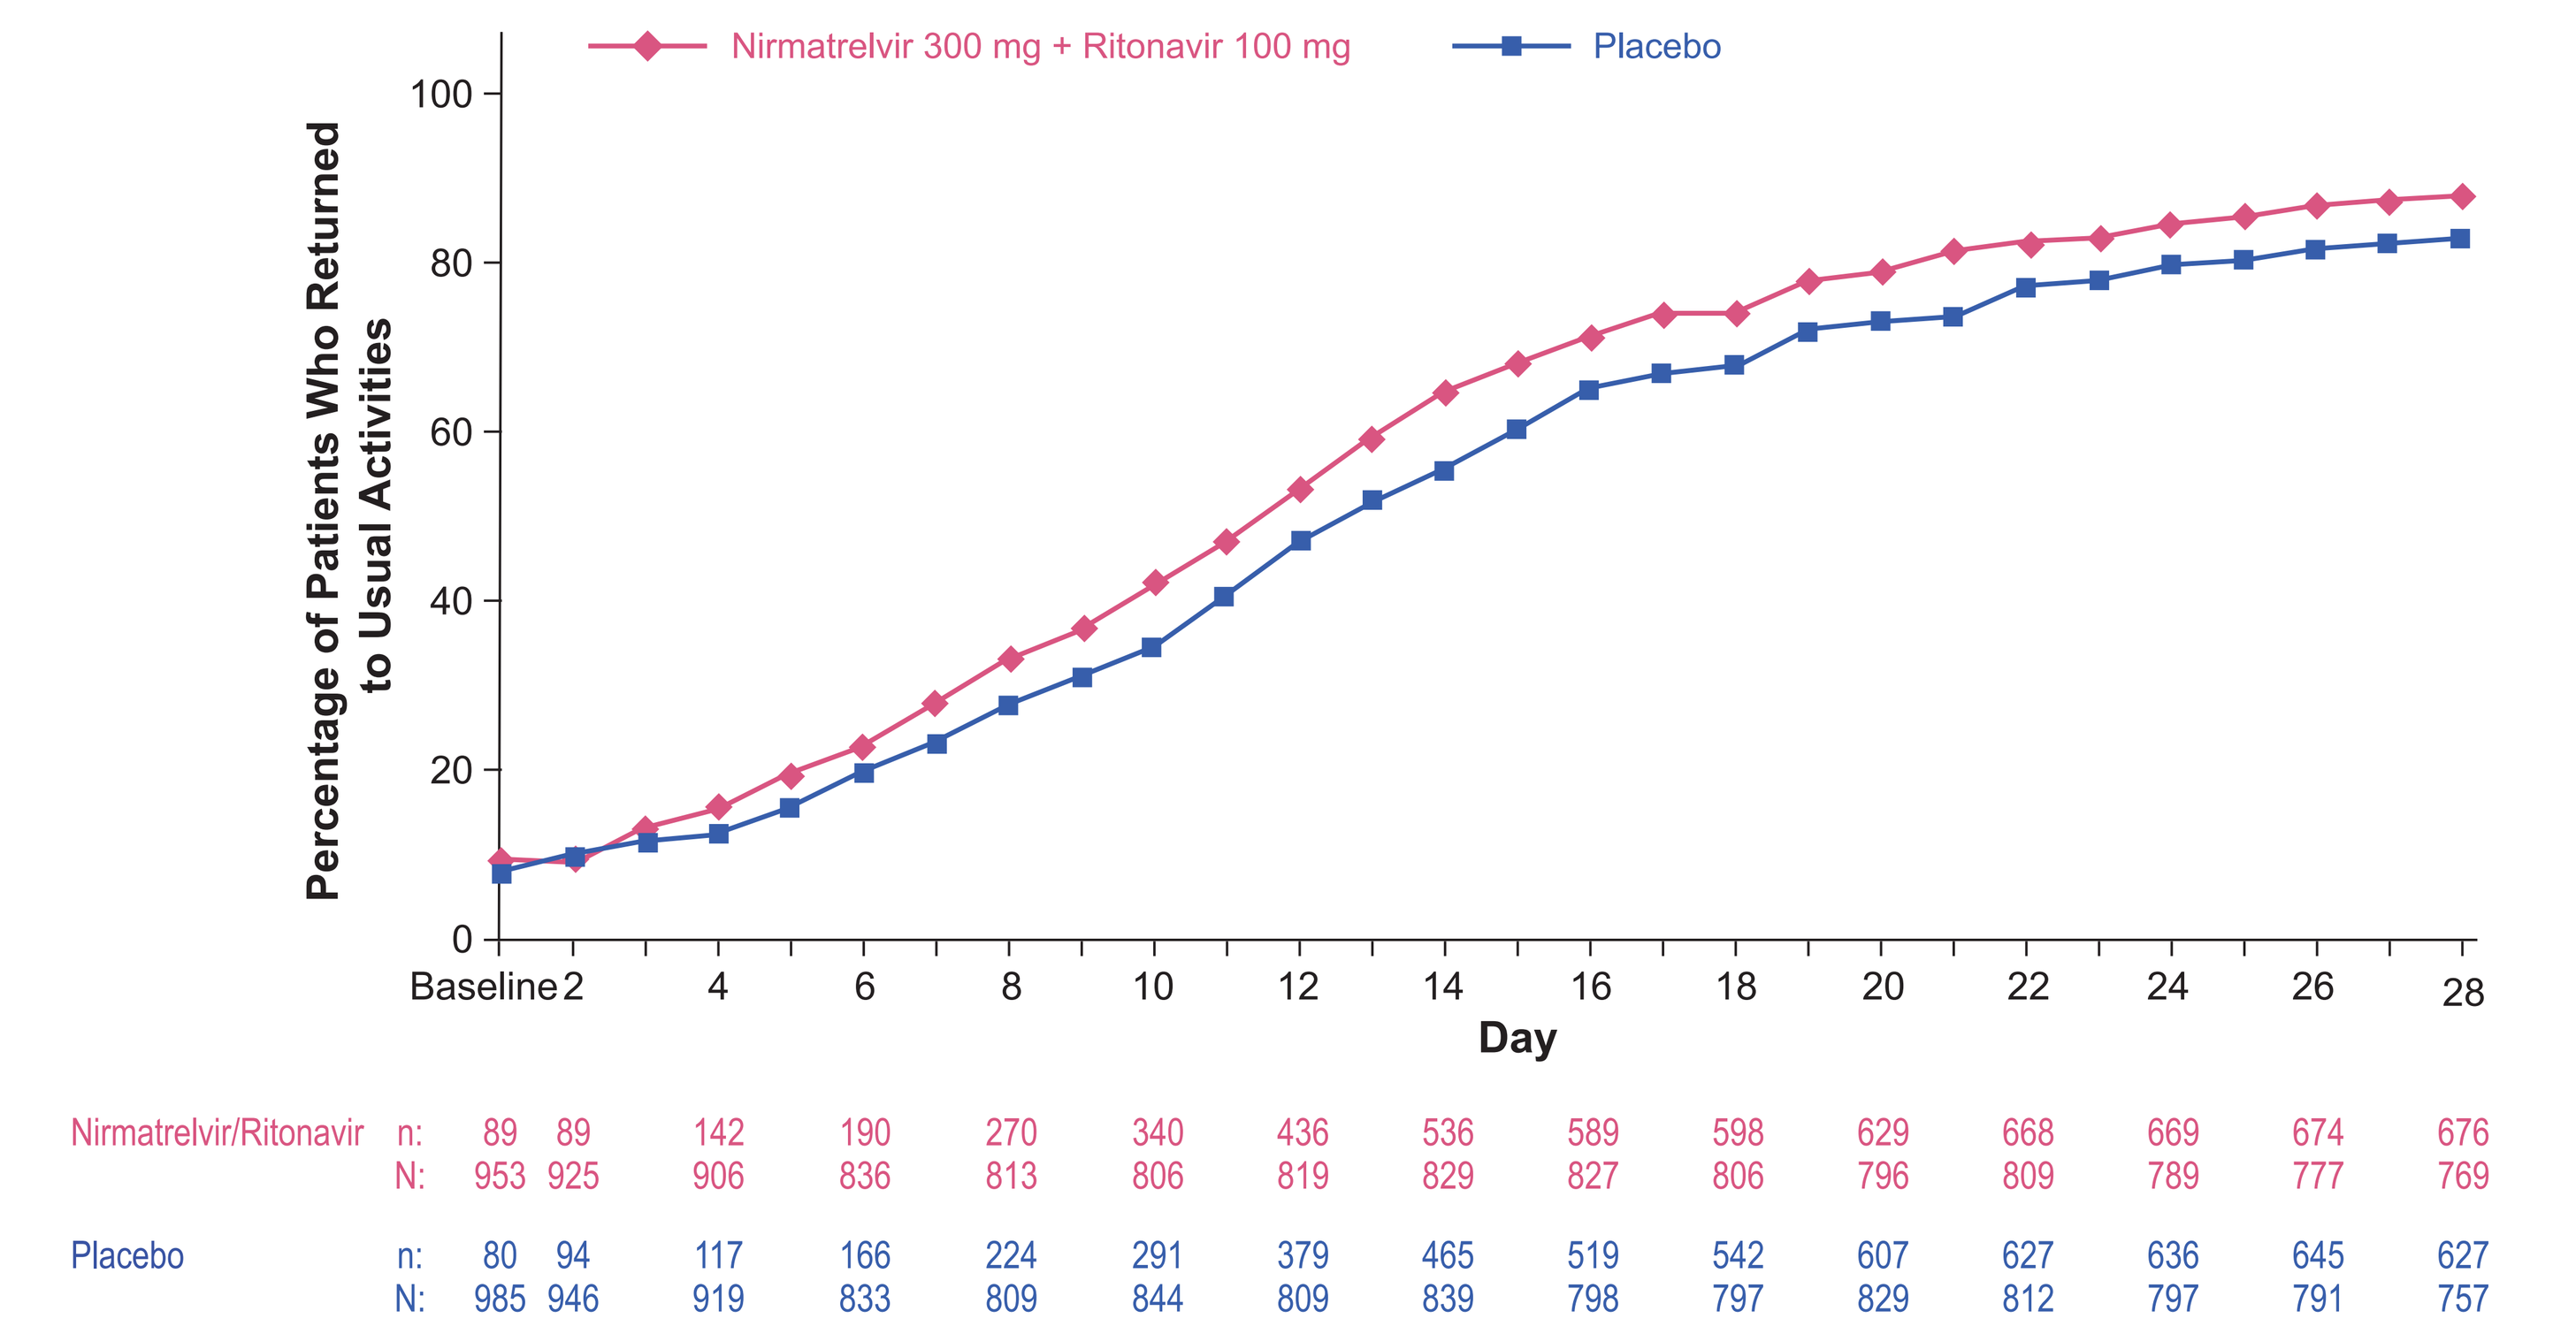
Nirmatrelvir 300 mg + Ritonavir 100 mg
Placebo
Percentage of Patients Who Returned to Usual Activities
Day
Baseline 2 4 6 8 10 12 14 16 18 20 22 24 26 28
Nirmatrelvir/Ritonavir n: 89 89 142 190 270 340 436 536 589 598 629 668 669 674 676
N: 953 925 906 836 813 806 819 829 827 806 796 809 789 777 769
Placebo n: 80 94 117 166 224 291 379 465 519 542 607 627 636 645 627
N: 985 946 919 833 809 844 809 839 798 797 829 812 797 791 757

mITT2=modified intent-to-treat 2; n=number of patients with event; N=number of patients with non-missing data.

- Compared with patients receiving placebo, patients who received NMV/r reported significantly reduced time to achieving sustained symptom resolution (HR, 1.2; 95% CI, 1.1–1.3; *P*=0.0002; **Figure 4A**) and sustained symptom alleviation (HR, 1.2; 95% CI, 1.1–1.3; *P*<0.0001; **Figure 4B**).
 - Median time to sustained symptom resolution was 15 days (95% CI, 14-16 days) among patients who received NMV/r and 19 days (95% CI, 17-20 days) among patients who received placebo.
 - Median time to sustained symptom alleviation was 8 days (95% CI, 7-9 days) among patients who received NMV/r and 10 days (95% CI, 9-11 days) among patients who received placebo.
 - By Day 3, 1.4% of patients receiving NMV/r and 3.1% of patients receiving placebo reported severe overall COVID-19 symptoms; except for Day 7, these differences persisted through Day 12.
- Because of the operational challenges in the rollout of the electronic diary for PROs and a low number of baseline responses to both the WPAI-COVID-19 (n=53) and EQ-5D-5L (n=51), no meaningful conclusions could be drawn regarding the impact of NMV/r treatment on work productivity, activity impairment, or overall HRQoL.

Figure 4. Time to Achieving (A) Sustained Resolution and (B) Alleviation of Any Overall Symptoms, According to the Global Impressions Questionnaire in the mITT2 Population

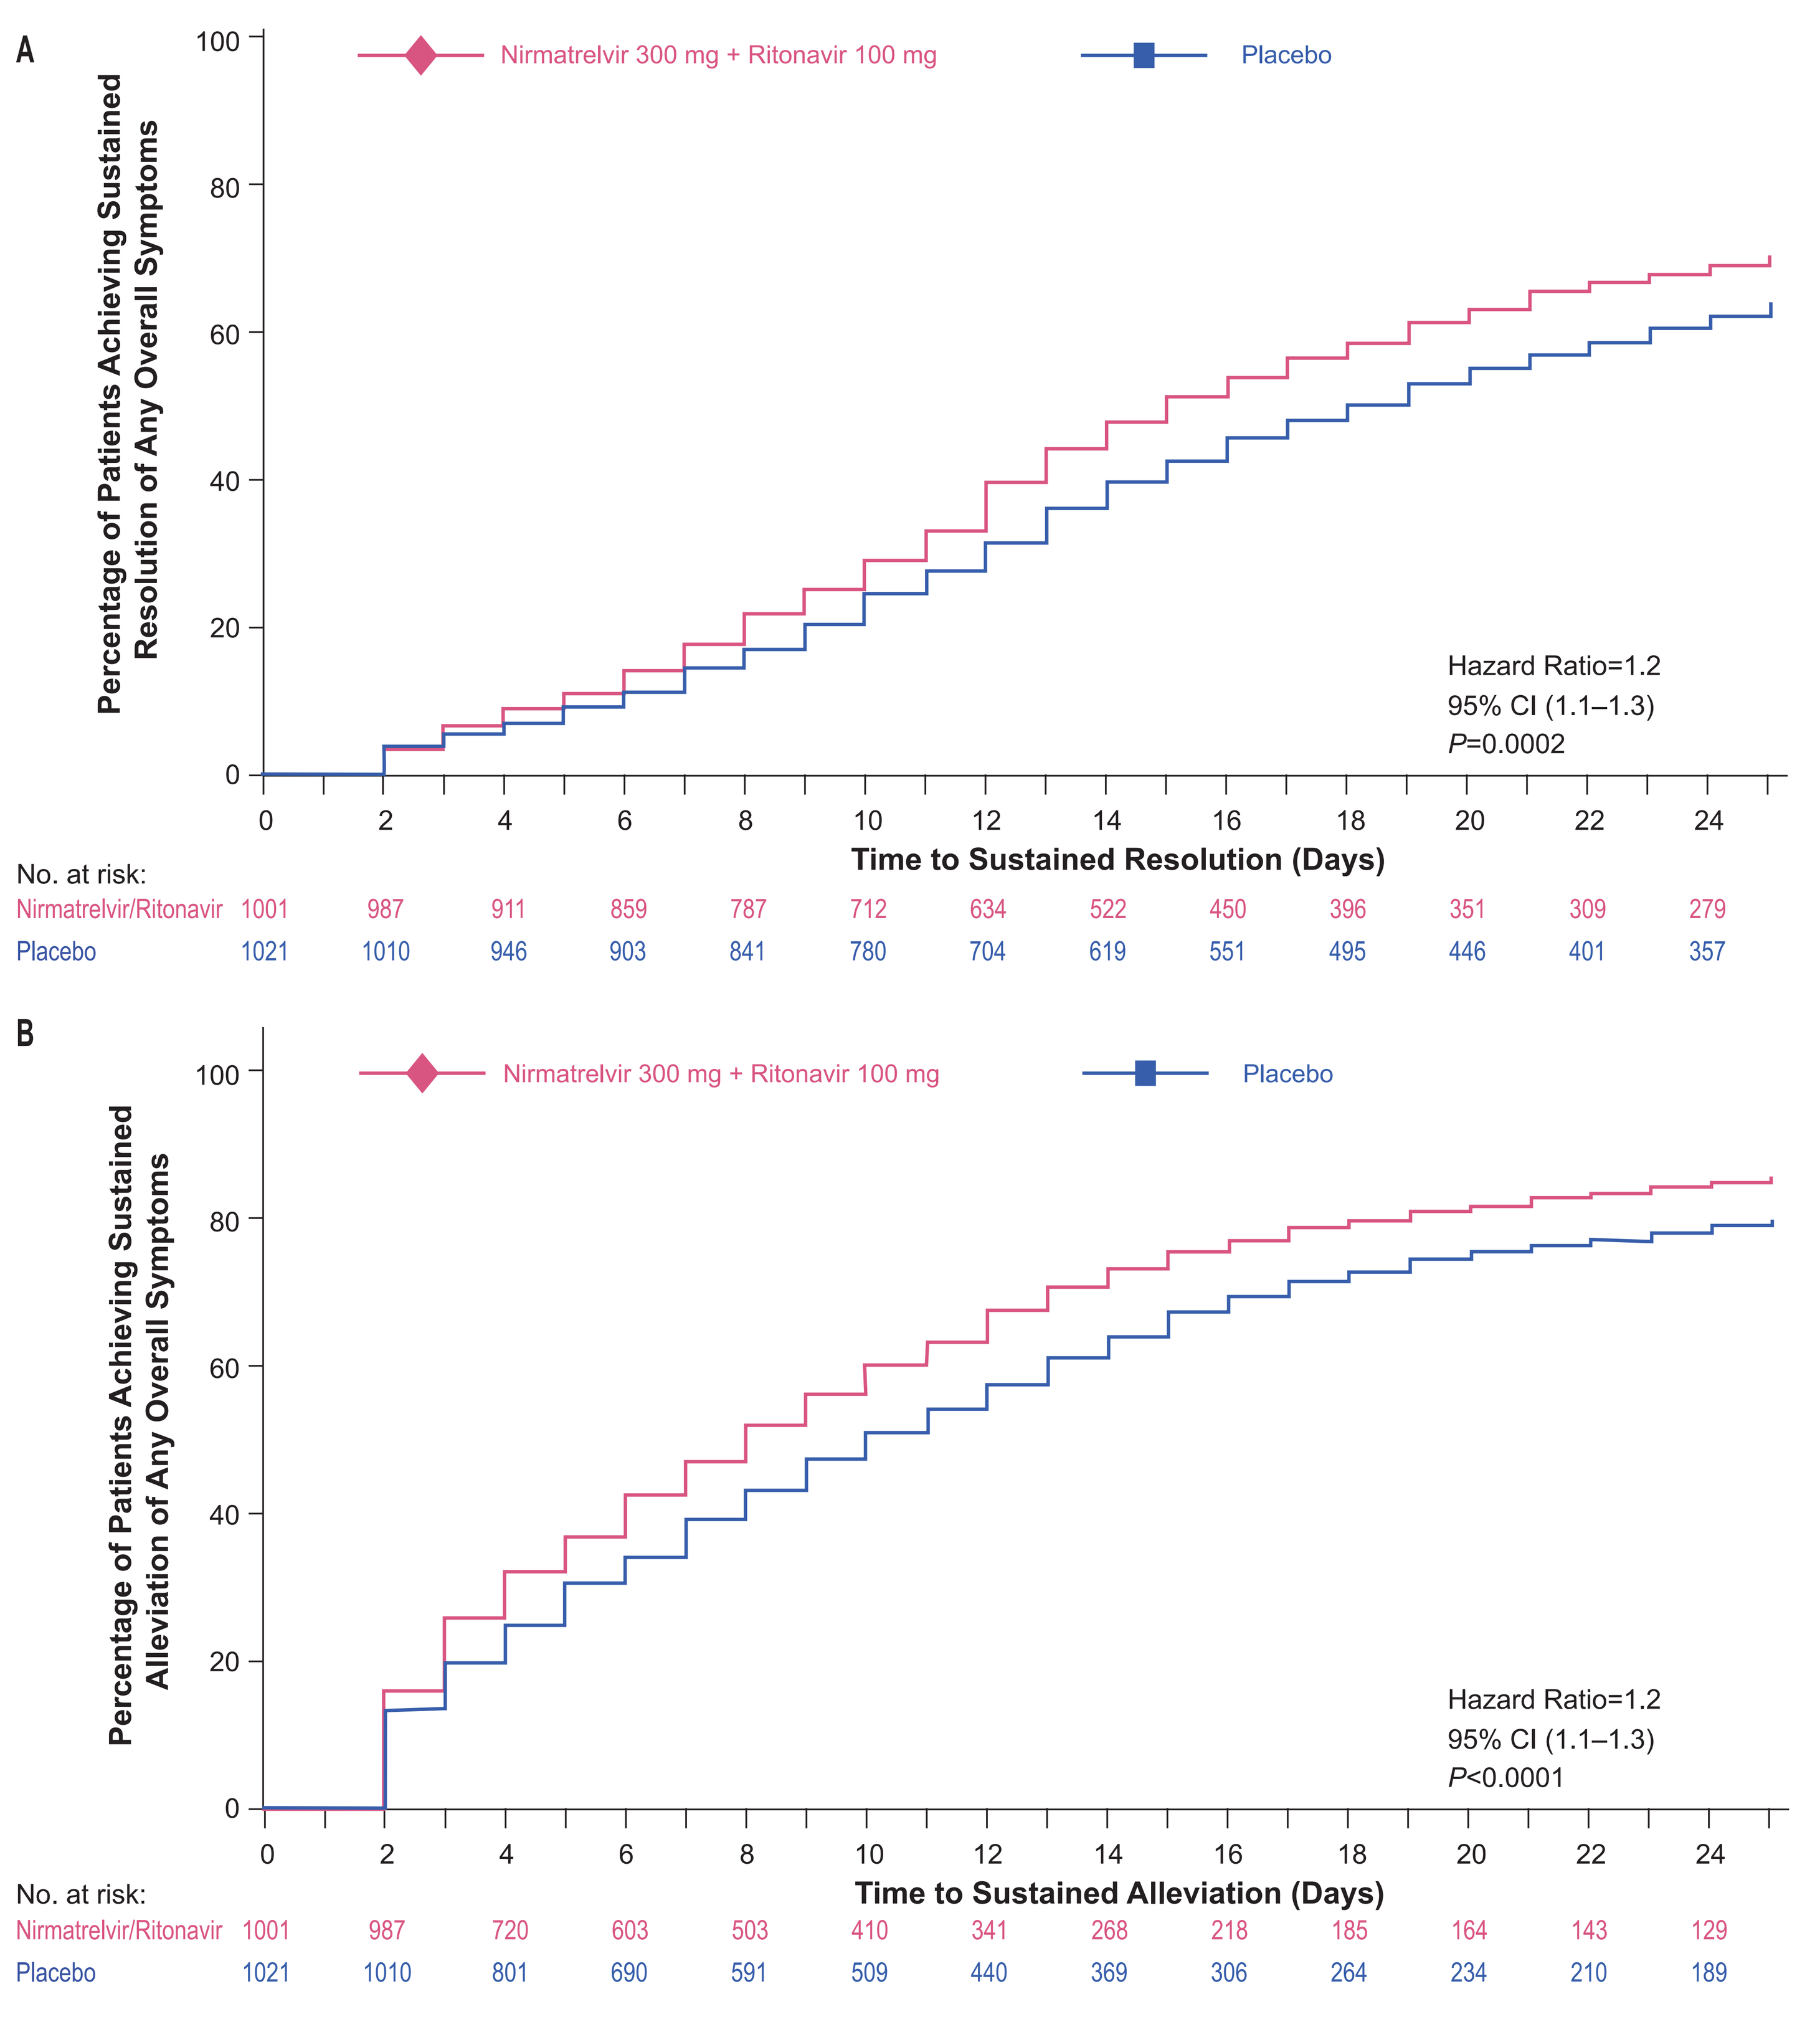
A
Nirmatrelvir 300 mg + Ritonavir 100 mg
Placebo
Percentage of Patients Achieving Sustained Resolution of Any Overall Symptoms
Hazard Ratio=1.2
95% CI (1.1–1.3)
P=0.0002
Time to Sustained Resolution (Days)
No. at risk:
Nirmatrelvir/Ritonavir 1001 987 911 859 787 712 634 522 450 396 351 309 279
Placebo 1021 1010 946 903 841 780 704 619 551 495 446 401 357
B
Nirmatrelvir 300 mg + Ritonavir 100 mg
Placebo
Percentage of Patients Achieving Sustained Alleviation of Any Overall Symptoms
Hazard Ratio=1.2
95% CI (1.1–1.3)
P<0.0001
Time to Sustained Alleviation (Days)
No. at risk:
Nirmatrelvir/Ritonavir 1001 987 720 603 503 410 341 268 218 185 164 143 129
Placebo 1021 1010 801 690 591 509 440 369 306 264 234 210 189

mITT2=modified intent-to-treat 2.

CONCLUSIONS

- **In EPIC-HR, patients who received NMV/r reported quicker return to usual health and activities, quicker resolution and alleviation of symptoms, and reduced overall COVID-19 symptom severity compared with those who received placebo.**
- **Consistent with the previously demonstrated efficacy of NMV/r in reducing hospitalization and death among COVID-19 patients at high risk of progression to severe disease,[10,15] results here confirm the positive impact of NMV/r on patient experiences after receiving treatment for COVID-19.**

References

1. Yamana TK, et al. *PLoS One.* 2023;18(4):e0275699.
2. Satapathy P, et al. *New Microbes New Infect.* 2024;57:101225.
3. World Health Organization. COVID-19 Epidemiological Update.
4. Amdal CD, et al. *Qual Life Res.* 2021;30(12):3367-3381.
5. Judy J, et al. *J Med Econ.* 2024;27(1):941-951.
6. Pfizer Inc. Pfizer's PAXLOVID Receives FDA Approval for Adult Patients at High Risk of Progression to Severe COVID-19.
7. Pfizer Inc. Highlights of Prescribing Information: PAXLOVID.
8. National Institutes of Health. Final Coronavirus Disease (COVID-19) Treatment Guidelines (February 29, 2024).
9. World Health Organization. Therapeutics and COVID-19. November 10, 2023.
10. Hammond J, et al. *N Engl J Med.* 2022;386(15):1397-1408.
11. Reilly MC, et al. *Pharmacoeconomics.* 1993;4(5):353-365.
12. EuroQol Research Foundation. EQ-5D-5L User Guide. 2019.
13. US Food and Drug Administration. Assessing COVID-19-Related Symptoms in Outpatient Adult and Adolescent Subjects in Clinical Trials of Drugs and Biological Products for COVID-19 Prevention or Treatment: Guidance for Industry.
14. Rosner B. *Fundamentals of Biostatistics.* 8th ed. Boston, MA: Cengage Learning; 2015.
15. Cha-Silva AS, et al. A systematic literature review evaluating real-world use of nirmatrelvir-ritonavir for the prevention of COVID-19-related hospitalization and death. Presented at: IDWeek, 2023; Boston, MA, USA.

Acknowledgments

These studies were sponsored by Pfizer Inc. Medical writing support was provided by Anna Stern, PhD, of ICON (Blue Bell, PA) and was funded by Pfizer. Authors would like to thank Paul Cislo for feedback and data oversight.

Disclosures

Ashley S. Cha-Silva, Jinma Ren, Amie Scott, Joseph C. Cappelleri, Wajeeha Ansari, and Heidi Leister-Tebbe are employees of Pfizer Inc and may hold stock or stock options. Henriette Coetzer and Kelly Gebo are unpaid advisors to Pfizer and have no competing interests to report.

For more information please contact:
Ashley S. Cha-Silva
Global Biopharmaceuticals Group
Pfizer Inc
66 Hudson Blvd E.
New York, NY, 10001 USA
Email: Ashley.Cha@pfizer.com

Presented at IDWeek 2024; October 16–19; Los Angeles, CA, USA