Abstract 8590

Impact of lorlatinib dose modifications on adverse event outcomes in the phase 3 CROWN study

Geoffrey Liu,[1] Ernest Nadal,[2] Shobhit Baijal,[3] Alessandra Bulotta,[4] Christina S. Baik,[5] Holger C. Thurm,[6] Anna Polli,[7] Makoto Nishio[8]

[1]Princess Margaret Cancer Centre, Toronto, ON, Canada; [2]Catalan Institute of Oncology, IDIBELL, L'Hospitalet, Spain; [3]University Hospitals Birmingham NHS Trust, Birmingham, UK; [4]San Raffaele Scientific Institute, Milan, Italy; [5]Fred Hutchinson Cancer Center, Seattle, WA, USA; [6]Pfizer, San Diego, CA, USA; [7]Pfizer, Milan, Italy; [8]Cancer Institute Hospital of Japanese Foundation for Cancer Research, Tokyo, Japan

Conclusions

- In the phase 3 CROWN study, about one-third of the patients treated with lorlatinib had 1 or 2 dose reductions
- Median time to dose reduction to the 75-mg dose was 7.1 months, and median time to dose reduction to the 50-mg dose was 11.3 months
- Dose reductions enabled patients to continue treatment with a median duration post reduction of 42.2 months for the duration on 75-mg dose and 20.7 months for the duration on 50-mg dose
- This post hoc analysis showed that dose reductions were effective in managing AEs associated with lorlatinib, with most evaluable events partially or completely resolved with 1 or 2 dose reductions
- These findings show the importance of dose modifications to mitigate toxicity and continue lorlatinib treatment for prolonged periods of time in patients with advanced *ALK*-positive NSCLC

Electronic Poster

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster. If you don't have a smartphone, access the poster via the internet at: https://scientificpubs.congressposter.com/p/2ihnj94atxnjc9m0

Abstract Plain Language Summary

Please scan this QR code with your smartphone to view a plain language summary of the abstract. If you do not have a smartphone, access the summary via the internet at: https://scientificpubs.congressposter.com/pls/2ihnj94atxnjc9m0

Correspondence: Gregory Liu, Geoffrey.Liu@uhn.ca

References: 1. Lorbrena (lorlatinib). Prescribing information. Pfizer; 2024. Accessed March 19, 2025. http://labeling.pfizer.com/ShowLabeling.aspx?id=11140 2. Shaw AT, et al. *N Engl J Med*. 2020;383:2018-2029. 3. Solomon BJ, et al. *J Clin Oncol*. 2024;42:3400-3409.

Acknowledgments: The authors thank the participating patients and their families, investigators, sub-investigators, research nurses, study coordinators, and operations staff. During the preparation of this work, the authors used a genAI tool (3/27/24; Pfizer; GPT-4o) to develop the first draft. After using this tool, the authors reviewed and edited the content as needed and take full responsibility for the content of the publication. Editorial assistance was provided by Caitlin Cash, PhD, of Nucleus Global, and funded by Pfizer.

Presented at the American Society of Clinical Oncology Annual Meeting 2025 | May 30-June 3, 2025 | Chicago, IL, USA

Background

- Lorlatinib, a potent, brain-penetrant anaplastic lymphoma kinase (ALK) tyrosine kinase inhibitor, is indicated for the treatment of patients with *ALK*-positive metastatic non-small cell lung cancer (NSCLC)[1,2]
- Approval of lorlatinib in the first line was based on the phase 3 CROWN study (NCT03052608), which demonstrated significantly longer progression-free survival (PFS) and higher intracranial response with lorlatinib than crizotinib[2,3]
- After 5 years of follow-up, median PFS was not reached in the lorlatinib group, corresponding to the longest PFS for any single-agent molecular targeted treatment in advanced NSCLC[3]
 - Post hoc analyses showed that lorlatinib dose reductions within the first 16 weeks had no impact on PFS or time to intracranial progression
- These findings underscore the importance of dose modifications to mitigate toxicity and maintain long-term treatment efficacy
- This post hoc analysis aimed to further characterize lorlatinib dose reductions and their impact on safety and adverse event (AE) outcomes

Methods

- The CROWN study is an ongoing, international, open-label, randomized, phase 3 trial comparing lorlatinib vs crizotinib in patients with previously untreated *ALK*-positive advanced NSCLC [2,3]
 - Patients were randomized 1:1 to receive oral lorlatinib 100 mg once daily (QD) or crizotinib 250 mg twice daily
- The CROWN protocol allowed ≤2 lorlatinib dose reductions in 25-mg increments
- This analysis used data from the 5-year CROWN follow-up to further assess time to dose reduction as well as duration of treatment with reduced dose and its impact on AEs and outcomes associated with lorlatinib
- Data cutoff for this analysis was October 31, 2023

Results

- At 5 years of follow-up, 49 of 149 patients (33%) in the lorlatinib arm had ≥1 lorlatinib dose reduction (**Figure 1**)
 - 24 patients had 1 dose reduction to 75 mg QD; treatment was ongoing in 67% of those patients
 - 25 patients had 2 dose reductions (to 75 mg QD and then to 50 mg QD); treatment was ongoing in 40% of those patients
- Of the 49 patients who had ≥1 lorlatinib dose reduction, 45 had ≥1 dose interruption
 - Median duration of dose interruption was 1.2 months (range, 0.1-29.4)

Figure 1: Lorlatinib treatment discontinuations by dose reduction

49 Patients had ≥1 lorlatinib dose reduction

24 Patients had 1 dose reduction to 75 mg QD	25 Patients had 2 dose reductions (to 75 mg QD and then to 50 mg QD)
8 Patients discontinued treatment 3 Had AE 2 Had progressive disease 2 Withdrew consent 1 Had other reason	15 Patients discontinued treatment 5 Had AE 5 Had progressive disease 4 Withdrew consent 1 Had died
At data cutoff: 16 Patients continued to receive lorlatinib	At data cutoff: 10 Patients continued to receive lorlatinib

AE, adverse event; QD, once daily.

- In patients who had 1 dose reduction (**Figure 2A**):
 - Median time to dose reduction was 7.1 months (range, 1.7-64.8)
 - Median duration of treatment post reduction with the 75-mg dose was 42.2 months (range, 0.2-68.3)
- In patients who had 2 dose reductions (**Figure 2B**):
 - Median time from baseline to second dose reduction was 11.3 months (range, 2.5-56.9)
 - Median duration of treatment post second reduction with the 50-mg dose was 20.7 months (range, 0.5-61.8)

Figure 2. Duration of treatment by dose reduction

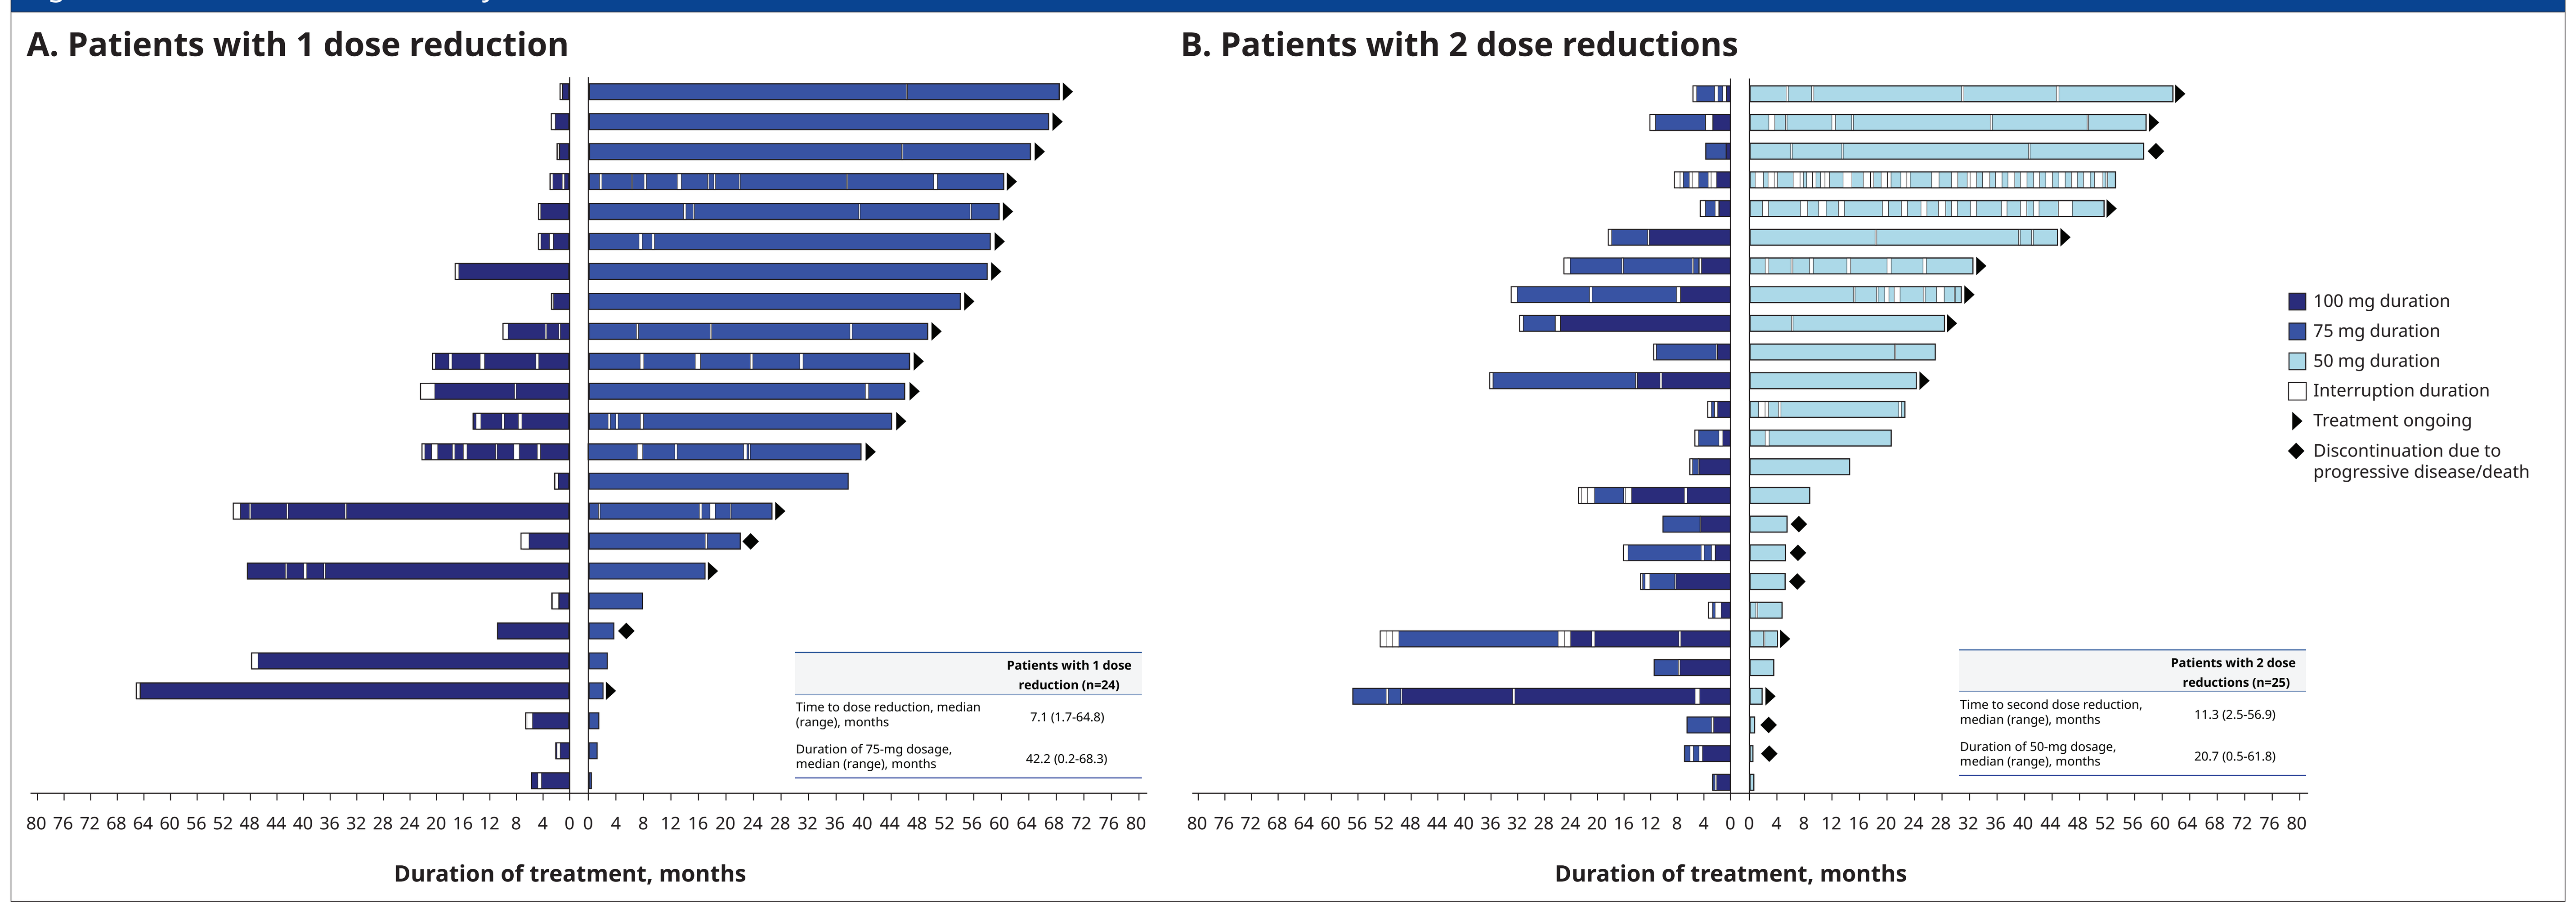

A. Patients with 1 dose reduction
B. Patients with 2 dose reductions
100 mg duration
75 mg duration
50 mg duration
Interruption duration
Treatment ongoing
Discontinuation due to progressive disease/death
Patients with 1 dose reduction (n=24)
Time to dose reduction, median (range), months 7.1 (1.7-64.8)
Duration of 75-mg dosage, median (range), months 42.2 (0.2-68.3)
Patients with 2 dose reductions (n=25)
Time to second dose reduction, median (range), months 11.3 (2.5-56.9)
Duration of 50-mg dosage, median (range), months 20.7 (0.5-61.8)
80 76 72 68 64 60 56 52 48 44 40 36 32 28 24 20 16 12 8 4 0 0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80
Duration of treatment, months

- In patients who had 1 or 2 dose reductions, peripheral edema was the most common all-cause AE associated with dose reductions (**Table 1**)
- 30 AE occurrences were associated with 1 dose reduction, and 59 were associated with 2 dose reductions (**Table 2**)
 - For 14 AE occurrences associated with 1 dose reduction and 21 AE occurrences associated with 2 dose reductions, outcomes following dose reduction could not be evaluated as the occurrences resolved with dose interruption prior to dose reduction
- Of the 16 evaluable AE occurrences associated with 1 dose reduction, 50% resolved and 25% partially resolved (**Figure 3**)
- Of the 38 evaluable AE occurrences associated with 2 dose reductions, 71% resolved and 8% partially resolved

Table 1: All-cause AEs associated with lorlatinib dose reductions

AE associated with dose reductions in ≥2 patients, n (%)	Any grade	Grade ≥3
1 dose reduction (n=24)		
Any	23 (96)	14 (58)
Peripheral edema	4 (17)	2 (8)
Alanine aminotransferase increased	2 (8)	0
Hypertriglyceridemia	2 (8)	1 (4)
2 dose reductions (n=25)		
Any	24 (96)	11 (44)
Peripheral edema	6 (24)	0
Blood triglycerides increased	3 (12)	2 (8)
Disturbance in attention	3 (12)	0
Generalized edema	3 (12)	1 (4)
Dysarthria	2 (8)	0
Gamma-glutamyltransferase increased	2 (8)	1 (4)
Hallucination	2 (8)	0
Hypercholesterolemia	2 (8)	0
Hypertriglyceridemia	2 (8)	0
Edema	2 (8)	0
Paresthesia	2 (8)	0
Weight increased	2 (8)	2 (8)

AE, adverse event.

Table 2: Summary of all-cause AE outcomes following dose reduction

	Outcome[a]			
AE with ≥2 occurrences, n (%)	Resolved	Partially resolved	Not resolved	Not applicable
AE occurrences associated with 1 dose reduction (n=30)				
All AE occurrences	8 (27)	4 (13)	4 (13)	14 (47)
Peripheral edema	1 (3)	1 (3)	2 (7)	0
Alanine aminotransferase increased	2 (7)	0	0	0
Hypertriglyceridemia	0	1 (3)	0	1 (3)
AE occurrences associated with 2 dose reductions (n=59)				
All AE occurrences	27 (46)	3 (5)	8 (14)	21 (36)
Peripheral edema	1 (2)	2 (3)	1 (2)	2 (3)
Blood triglycerides increased	3 (5)	0	0	0
Disturbance in attention	0	0	1 (2)	2 (3)
Generalized edema	2 (3)	0	0	1 (2)
Dysarthria	1 (2)	0	0	1 (2)
Gamma-glutamyltransferase increased	0	0	0	2 (3)
Hallucination	1 (2)	0	0	1 (2)
Hypercholesterolemia	2 (3)	0	0	0
Hypertriglyceridemia	2 (3)	0	0	0
Edema	1 (2)	1 (2)	0	0
Paresthesia	1 (2)	0	1 (2)	0
Weight increased	1 (2)	0	1 (2)	0

AE, adverse event.

[a]Resolved was reported if the event resolved or returned to baseline. Partially resolved was reported if the event is not resolved but improved versus the worst grade observed. Not resolved was reported if the event does not fall in 1 of the 2 previous categories. Not applicable was reported if the event resolved before or on associated dose reduction start date.

Figure 3. Outcomes[a] of all-cause AE occurrences following dose reduction

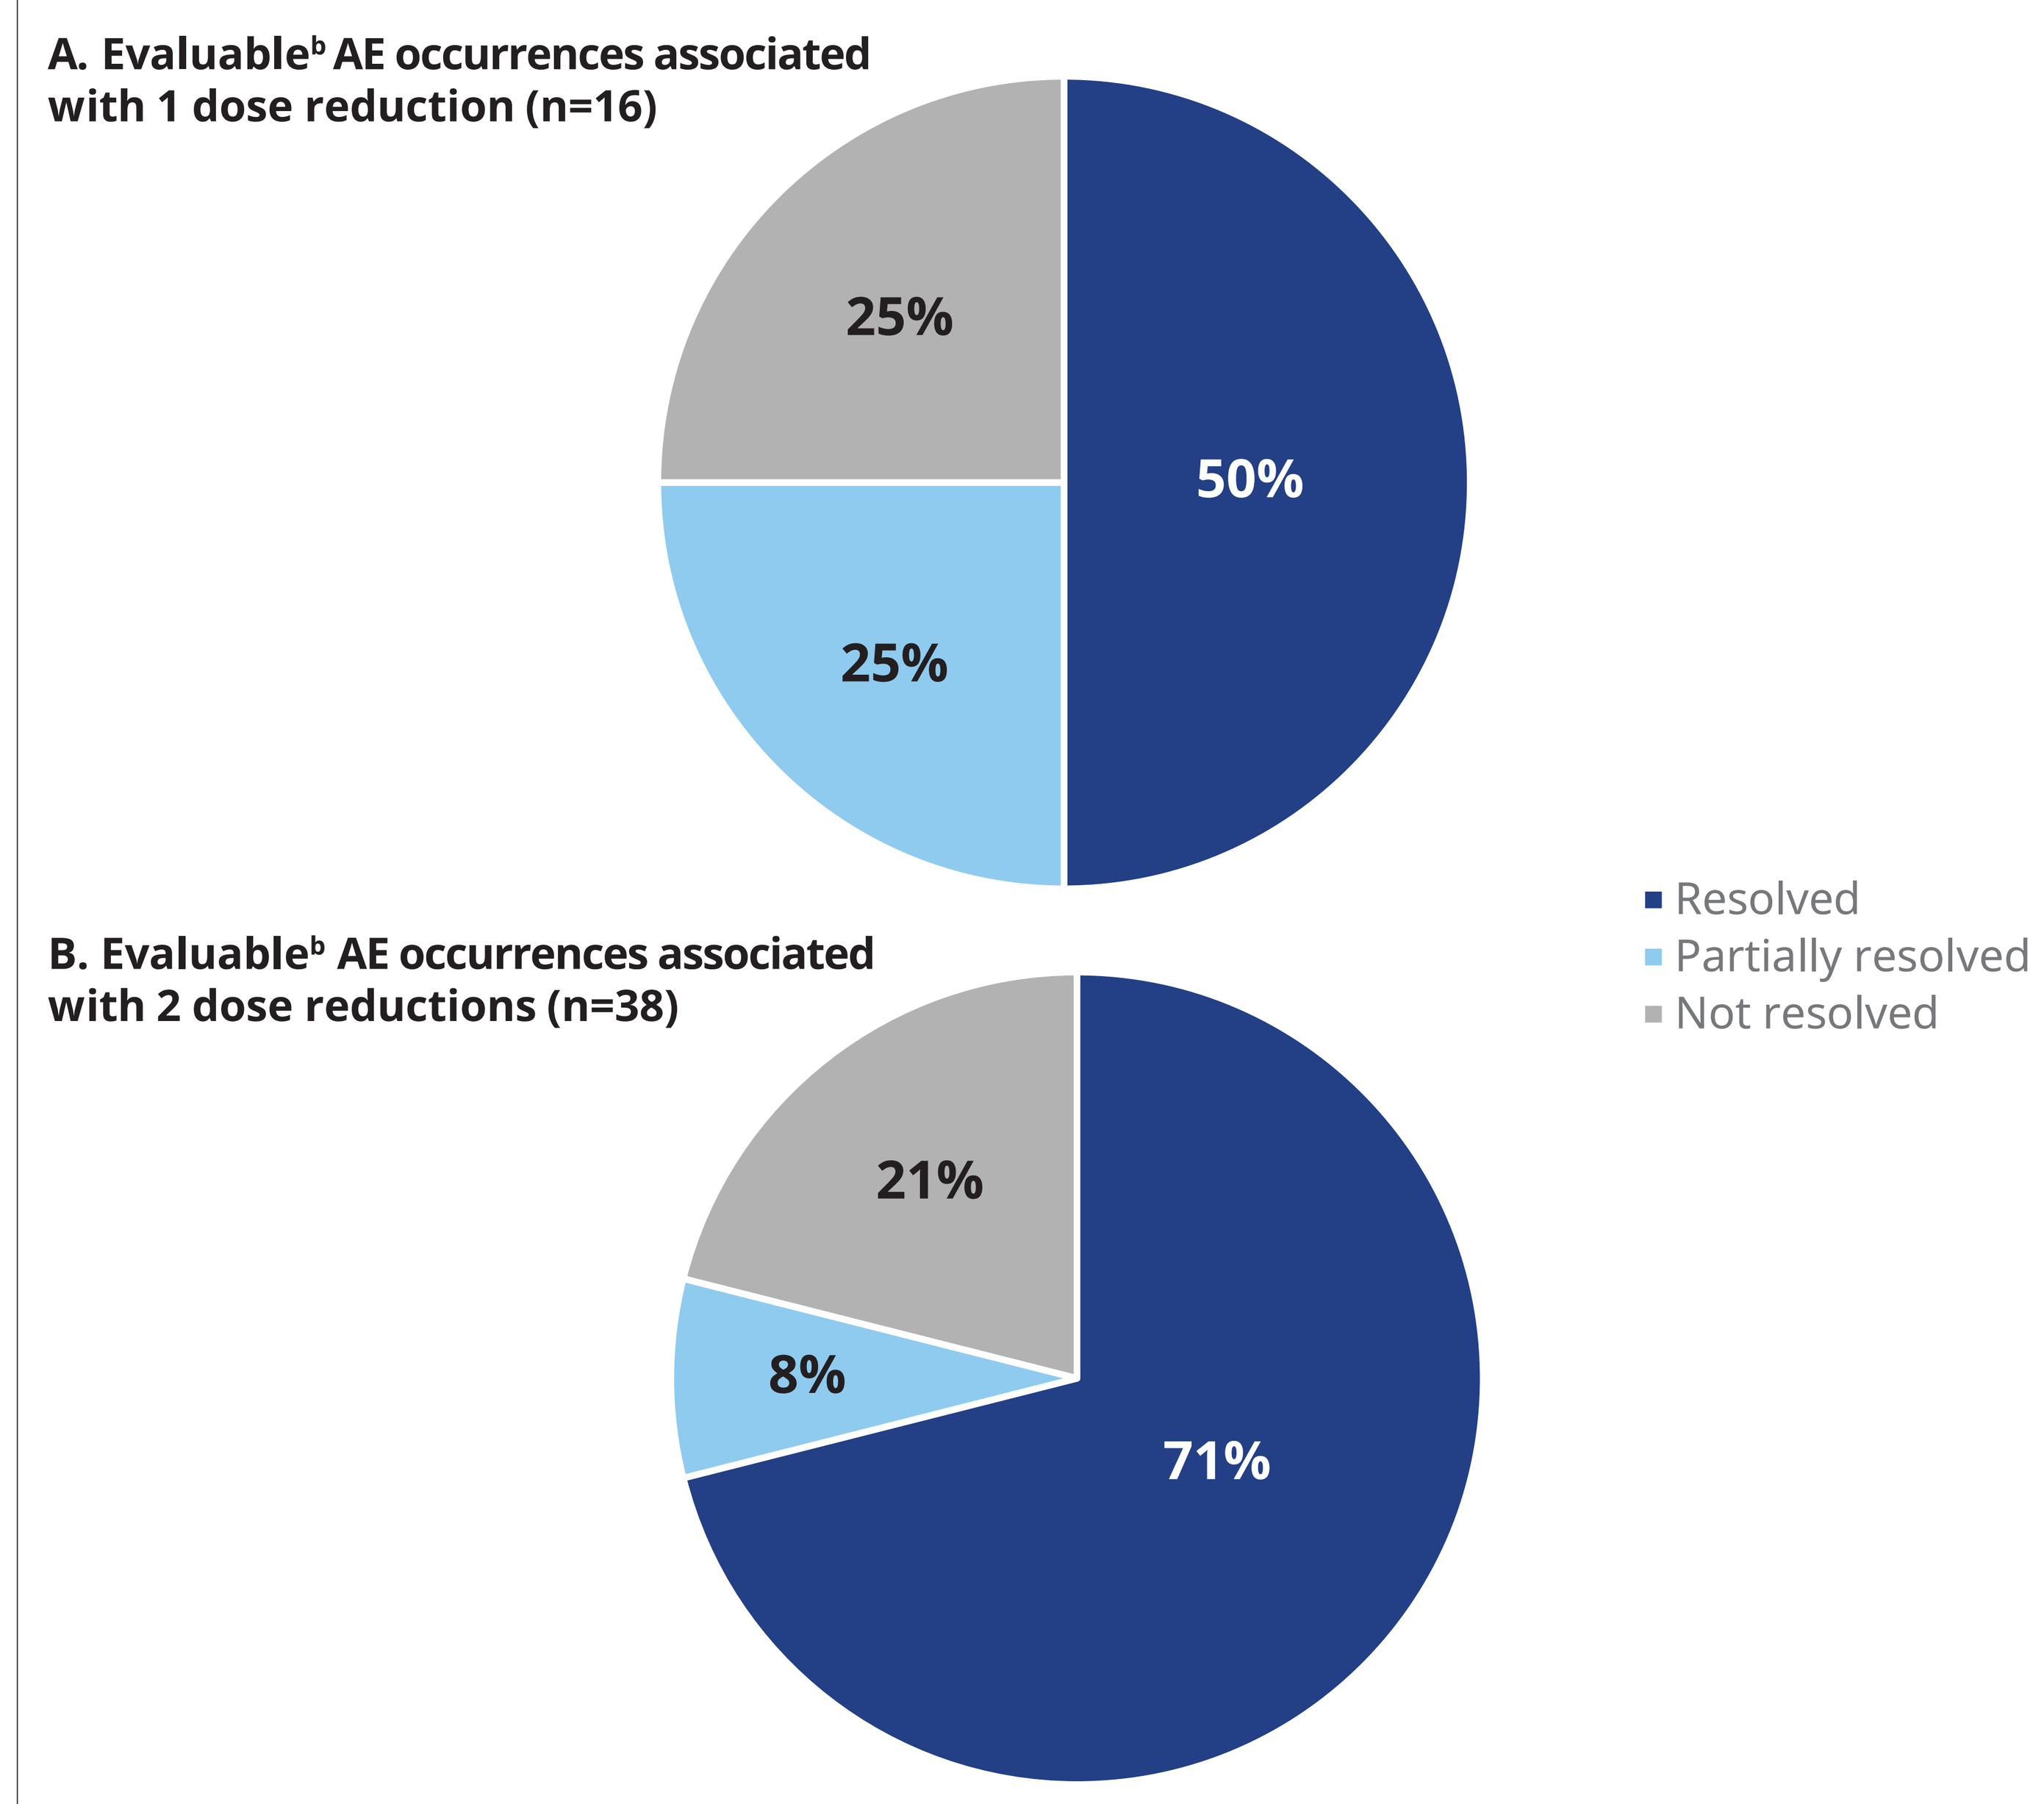

A. Evaluable[b] AE occurrences associated with 1 dose reduction (n=16)
50%
25%
25%
B. Evaluable[b] AE occurrences associated with 2 dose reductions (n=38)
71%
8%
21%
Resolved
Partially resolved
Not resolved

AE, adverse event.

[a]Resolved was reported if the event resolved or returned to baseline. Partially resolved was reported if the event is not resolved but improved vs the worst grade observed. Not resolved was reported if the event does not fall in 1 of the 2 previous categories. [b]Resolution was not evaluable for events that resolved before or on associated dose reduction start date.